Establishment of a ddPCR Method for Detecting Epstein-Barr Virus in MS Samples

Tassja J Spindler, Jenny Xiao, Jason Dubovsky, and Monica Moreno
Atara Biotherapeutics, Thousand Oaks, United States

BACKGROUND

- Accumulating evidence shows that Epstein Barr virus (EBV) infection, particularly in B cells, may be involved in the pathogenesis of multiple sclerosis (MS)(1-2)
 - EBV infection has been reported in up to 100% of MS patients(3-5)
 - Higher titers of antibodies to EBNA are associated with increased risk of developing MS(6)
 - MS risk increases sharply in individuals following EBV infection(7-8)
 - Increased prevalence of EBV-infected B cells in brain tissue(9-10)
 - Alterations in EBV-specific CD8+ T-cell immunity(11-12)
- ATA188 is an investigational off the shelf, allogeneic T-cell immunotherapy that targets EBV infected cells and has shown promising initial safety and efficacy (ECTRIMS 2021, ECF 2020)
 - Currently in Phase 2 clinical study for the treatment of progressive MS (EMBOLD Trial, NCT03283826)
- EBV is a member of the gamma herpesvirus subfamily with a genome size of 170kb that can live life-long in latency in the form of episomes in the host cells nucleus(13)
 - EBV DNA can be found cell associated in PBMCs, whole blood and tumor associates(14-16)
 - EBV DNA can also be found in cell free samples compartments as a result of replicating virus, apoptotic EBV+ cells (epithelial-, -B, NK-, T-cells)
- In healthy individuals it is estimated that 1-50 infected B cells per million B cells exist, while EBV associated diseases have 3 to 10-fold higher frequencies of infected B cells in the blood(17-18)
- A standardized method to quantify the presence of cell associated EBV DNA has not been developed
- Here we employ the highly sensitive droplet digital PCR (ddPCR) method for the purpose of testing EBV viral load in clinical PBMCs and CSF-derived cells from the EMBOLD study testing isolated genomic DNA

METHODS

- Natural sequence variation has been described at many locations in the genome of EBV isolates that lead to two different types of EBV (type 1 and type 2) as well as different strains within these types
- It is essential to design primers/probes to highly conserved target genes to provide robustness in detecting the various types and strains that may exist in cases and controls while remaining specific to EBV
- To optimize detection of varying EBV strains, we designed primer/probes specific to conserved EBV associated target genes BWRF1 and BCLF1
 - BCLF1 is a single copy gene
 - BWRF1 is a multi-repeat gene
- Selection of two gene targets integrates an internal confirmation of positivity and increased sensitivity
- In-silico testing was performed against a database of known viral genomes to confirm specificity to EBV
 - ViPR Virus Pathogen Resource

Figure 1.

Defective elimination of EBV-infected B cells by cytotoxic CD8+ T cells results in the accumulation of EBV-infected autoreactive B cells in lymphoid structures and within the CNS

1. Autoreactive naïve B cells in the tonsil are infected by EBV where they migrate to the germinal center (GC)
2. EBV-infected autoreactive B cells proliferate and differentiate into memory B cells which enter the circulation and eventually the CNS across the BBB
3. Autoreactive T cells cross the BBB into the CNS where they recruit other autoreactive T cells to the brain
4. **Possible Pathogenic Mechanism #1:** EBV-infected B cells entering the brain reside there for long periods of time where they may contribute to the production of oligoclonal IgG bands and pathogenic autoantibodies that attack and damage myelin sheath
5. **Possible Pathogenic Mechanism #2:**
 - Autoreactive T cells in the CNS, that would normally undergo apoptosis, receive survival signals from EBV-infected B cells (B7) through CD28 receptors.
 - Autoreactive T cells are reactivated by EBV-infected B cells presenting CNS antigens bound to MHC molecules.
 - Reactivated autoreactive T cells produce cytokines that recruit other inflammatory cells [effector and cytotoxic T cells] that drive an inflammatory cascade damaging oligodendrocytes, myelin, and neurons.

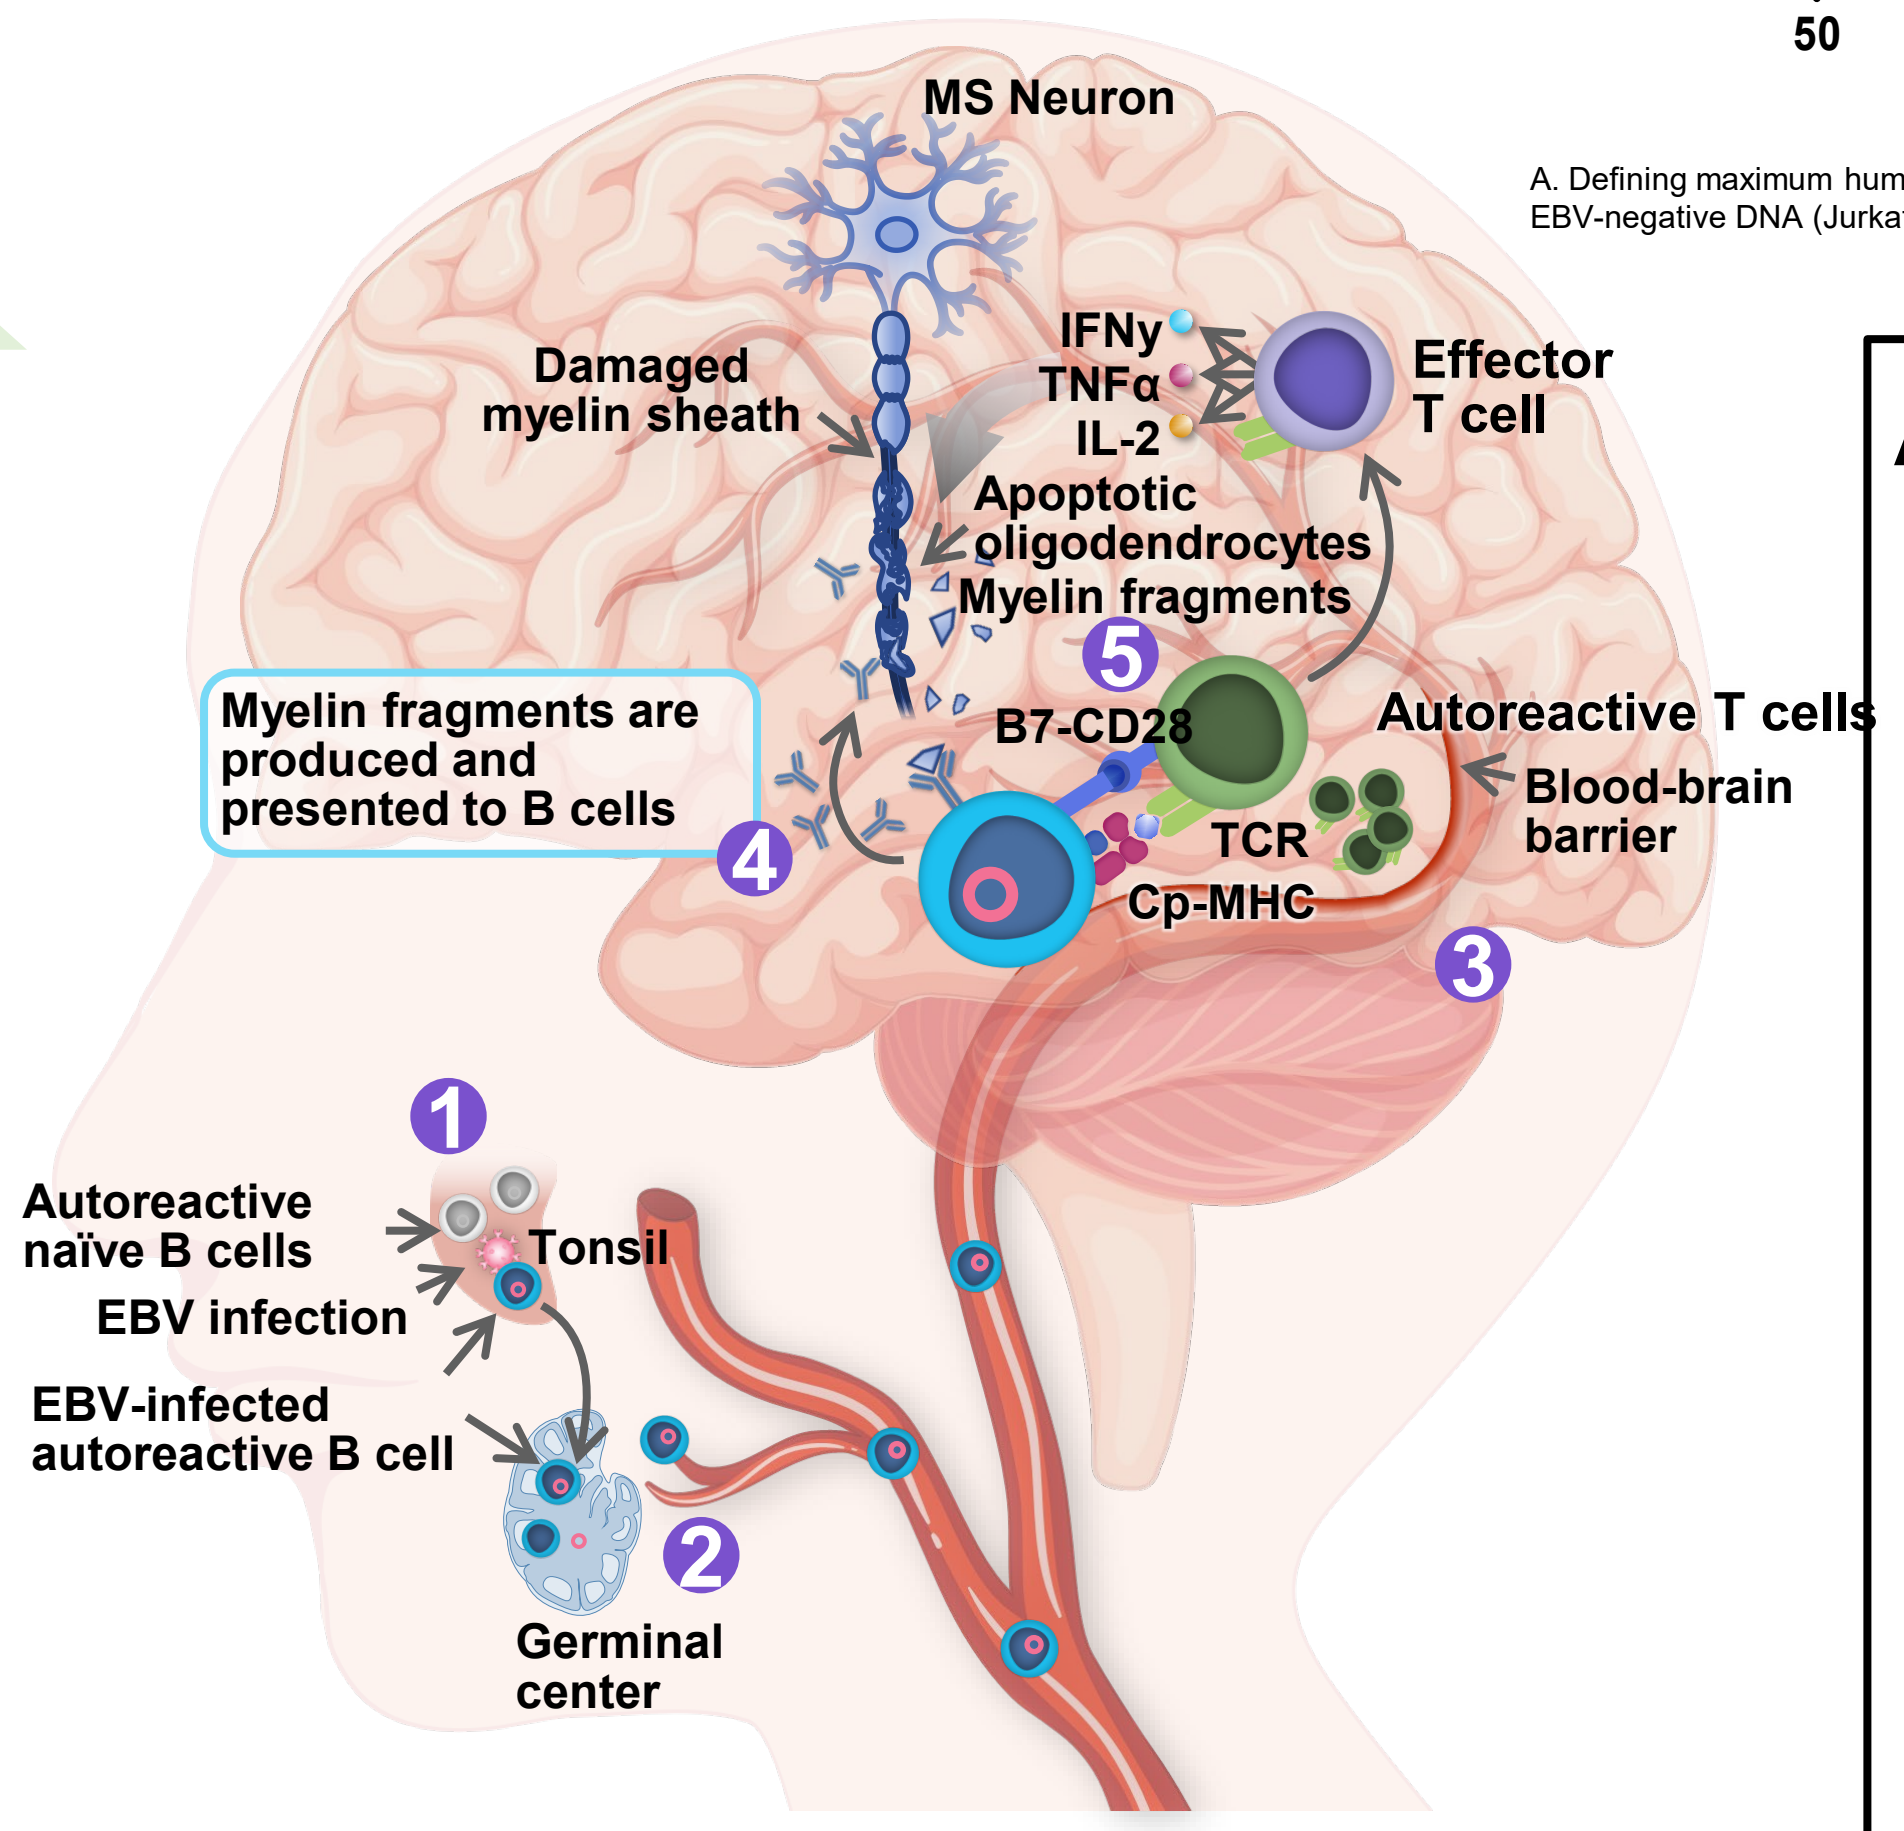
MS Neuron
Damaged myelin sheath
IFNy
TNFα
IL-2
Effector T cell
Apoptotic oligodendrocytes
Myelin fragments
Myelin fragments are produced and presented to B cells
B7-CD28
Autoreactive T cells
TCR
Cp-MHC
Blood-brain barrier
Autoreactive naïve B cells
Tonsil
EBV infection
EBV-infected autoreactive B cell
Germinal center

Figure 2. Viral Genome and Sample Collection Compartments

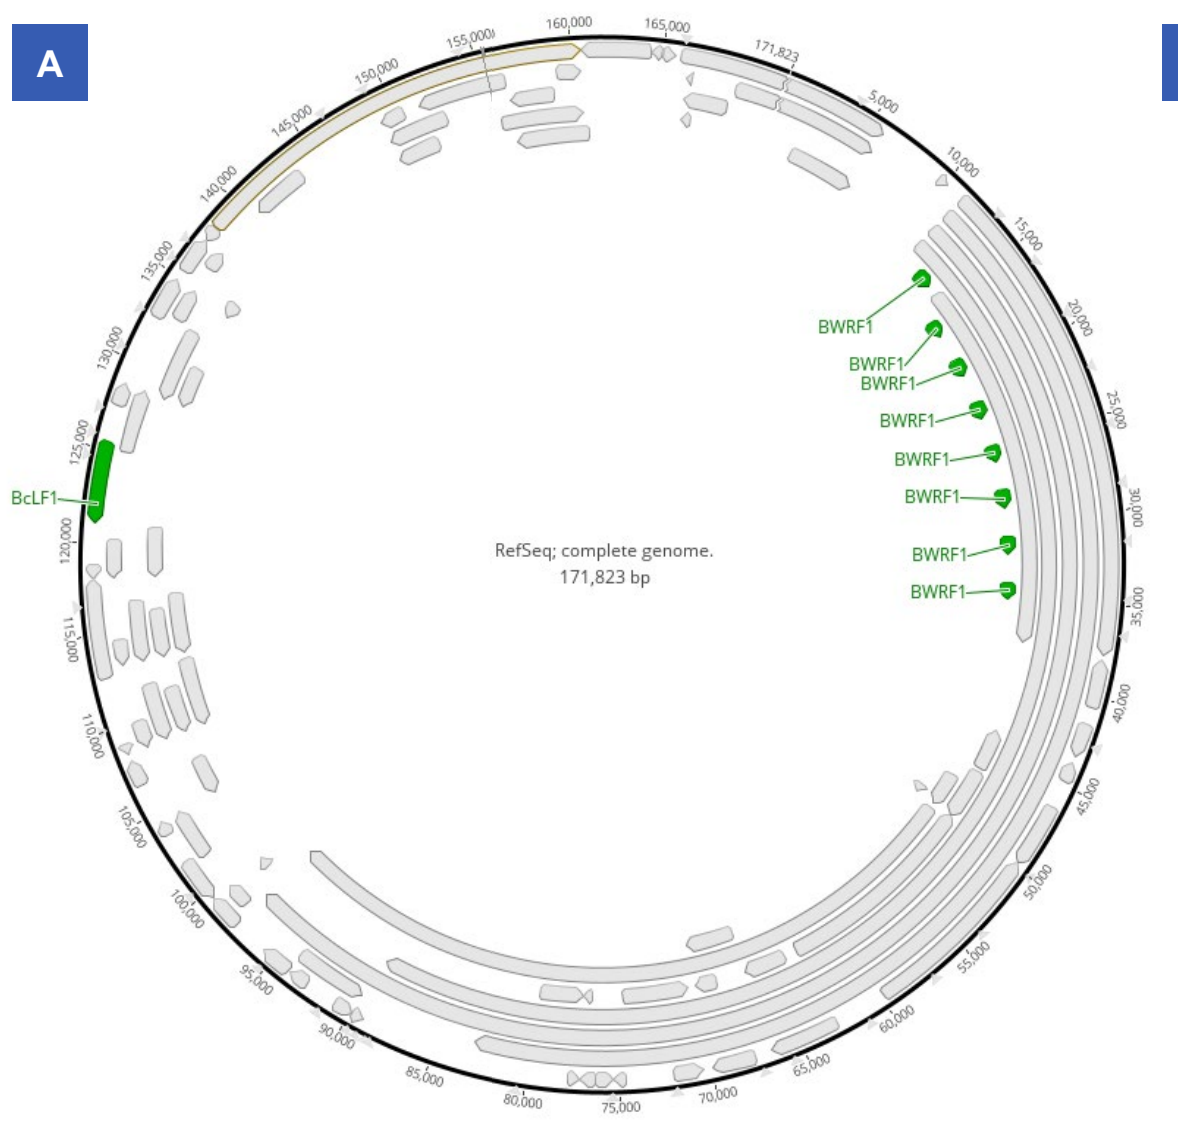
A
BcLF1
BWRF1
RefSeq; complete genome.
171,823 bp

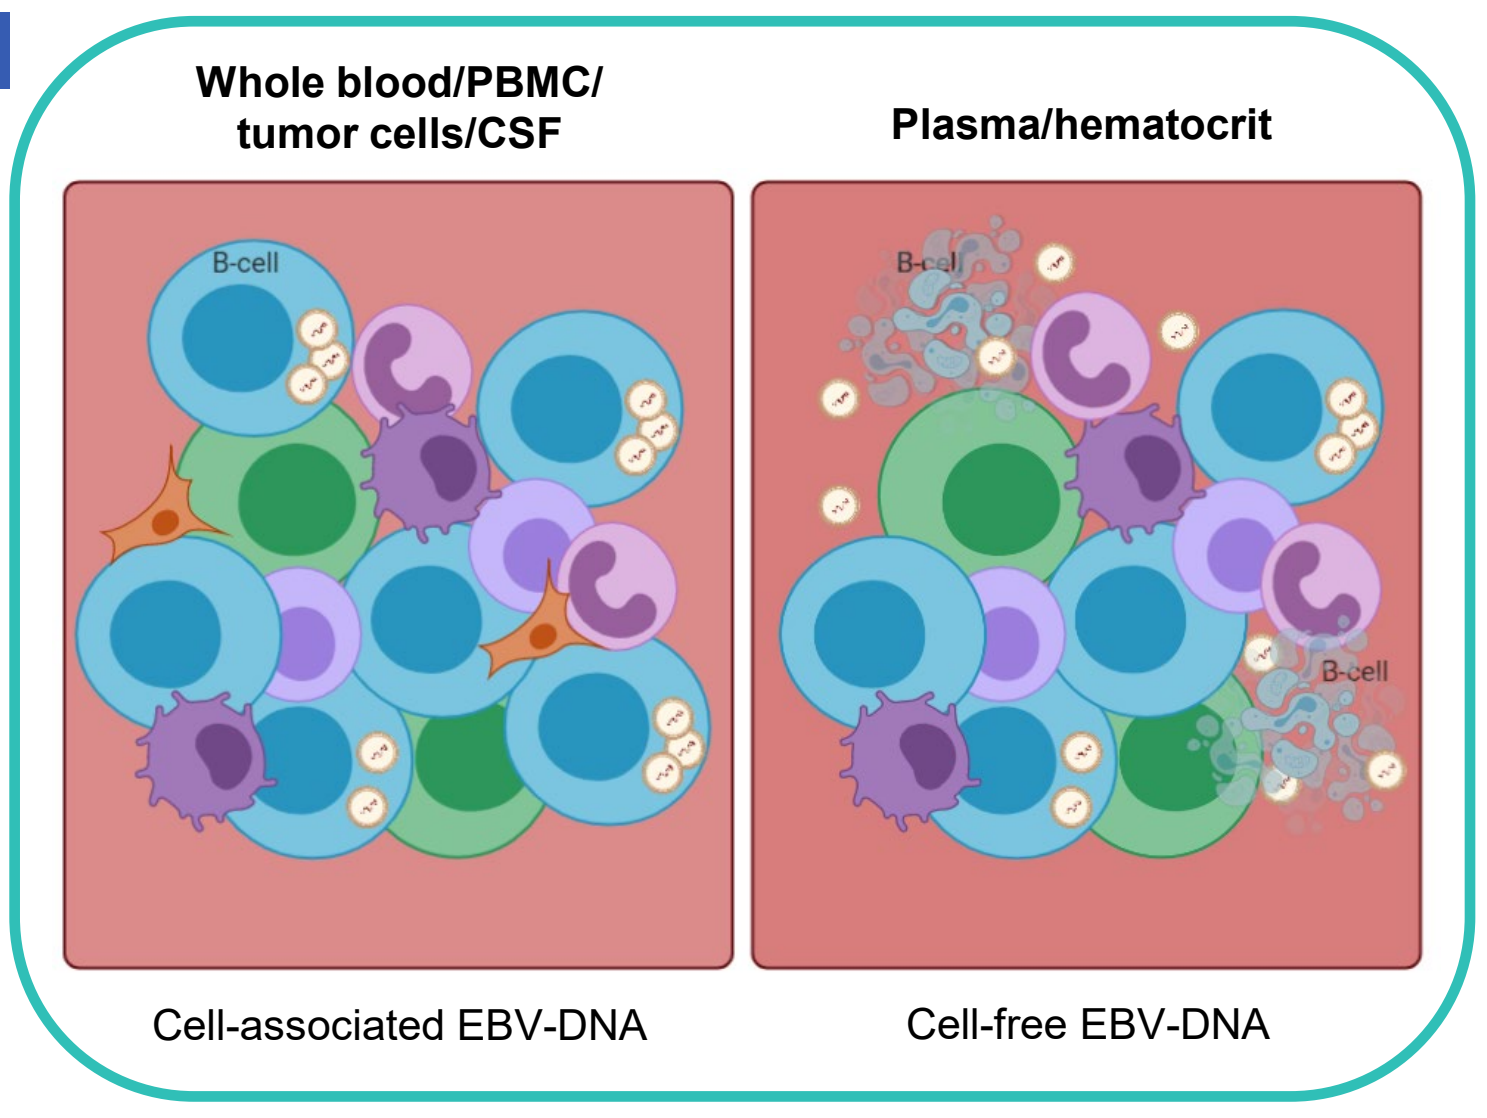
B
Whole blood/PBMC/ tumor cells/CSF
Plasma/hematocrit
B-cell
Cell-associated EBV-DNA
Cell-free EBV-DNA

A. Epstein-Barr Virus reference genome B. EBV-DNA sample compartments.

Figure 3. Method Workflow

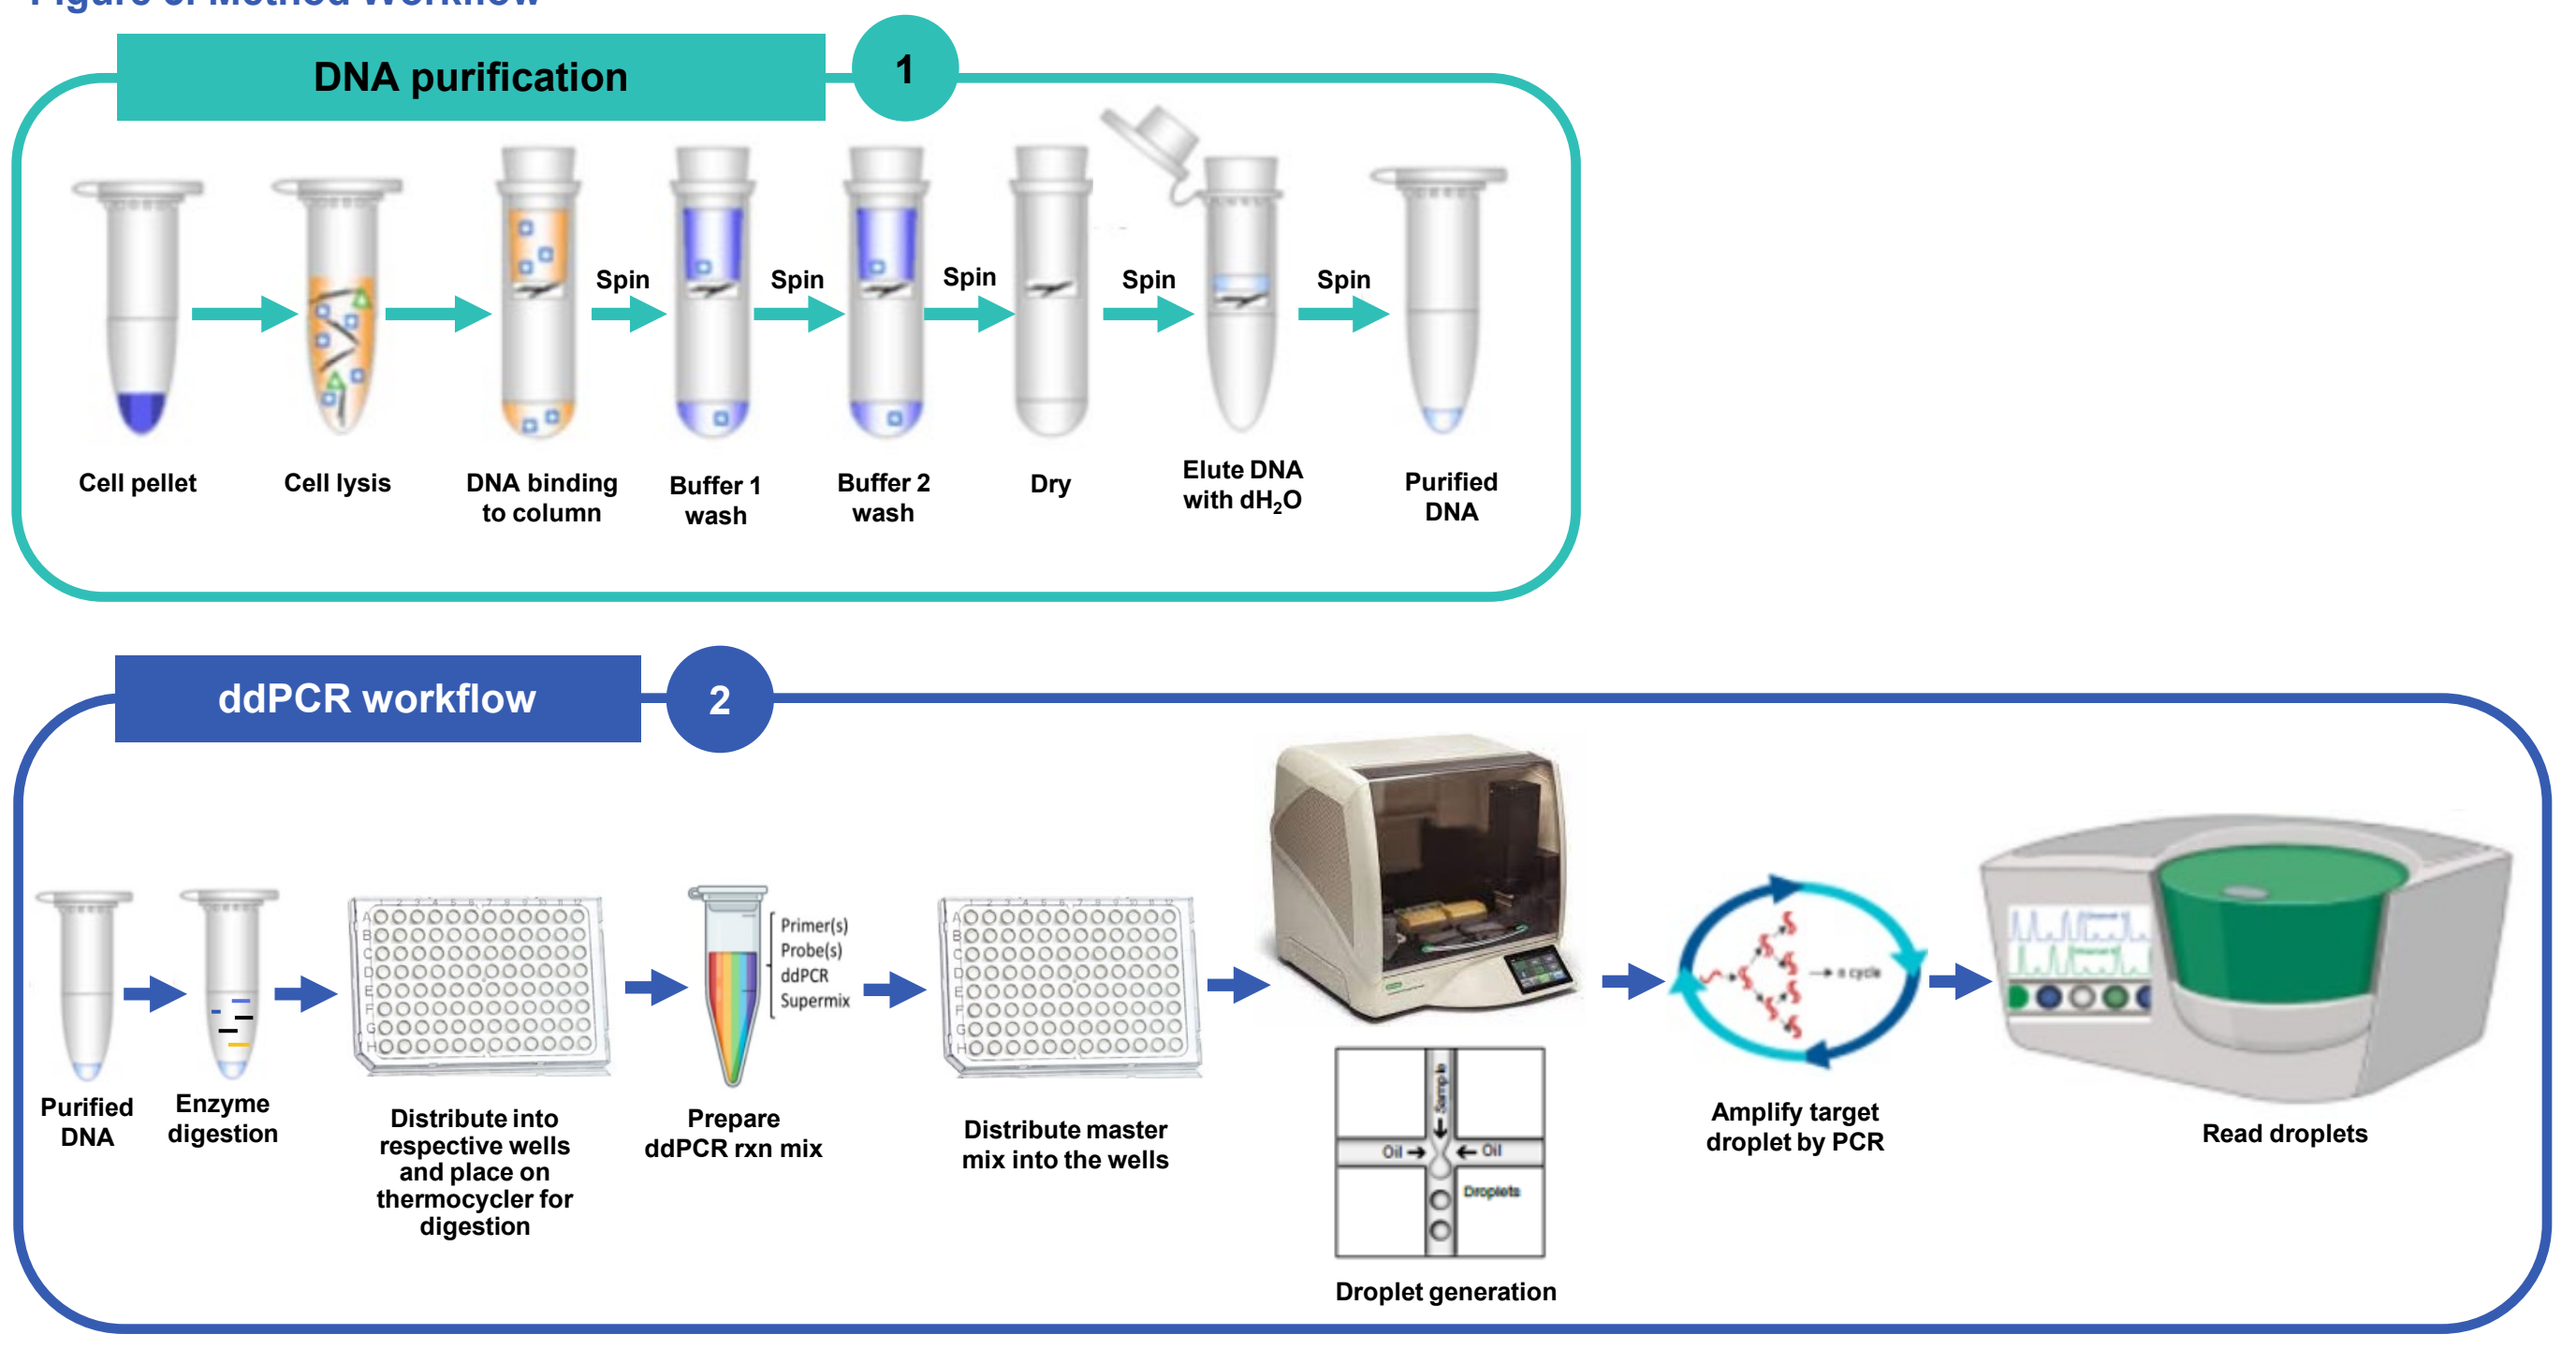
DNA purification
1
Cell pellet
Cell lysis
DNA binding to column
Spin
Buffer 1 wash
Buffer 2 wash
Dry
Elute DNA with dH2O
Purified DNA
ddPCR workflow
2
Purified DNA
Enzyme digestion
Distribute into respective wells and place on thermocycler for digestion
Primer(s)
Probe(s)
ddPCR
Supermix
Prepare ddPCR rxn mix
Distribute master mix into the wells
Oil
Droplets
Droplet generation
Amplify target droplet by PCR
Read droplets

Figure 4. BWRF1 and BCLF1 Gene Targets Exhibit Specificity to EBV+ Genomic DNA

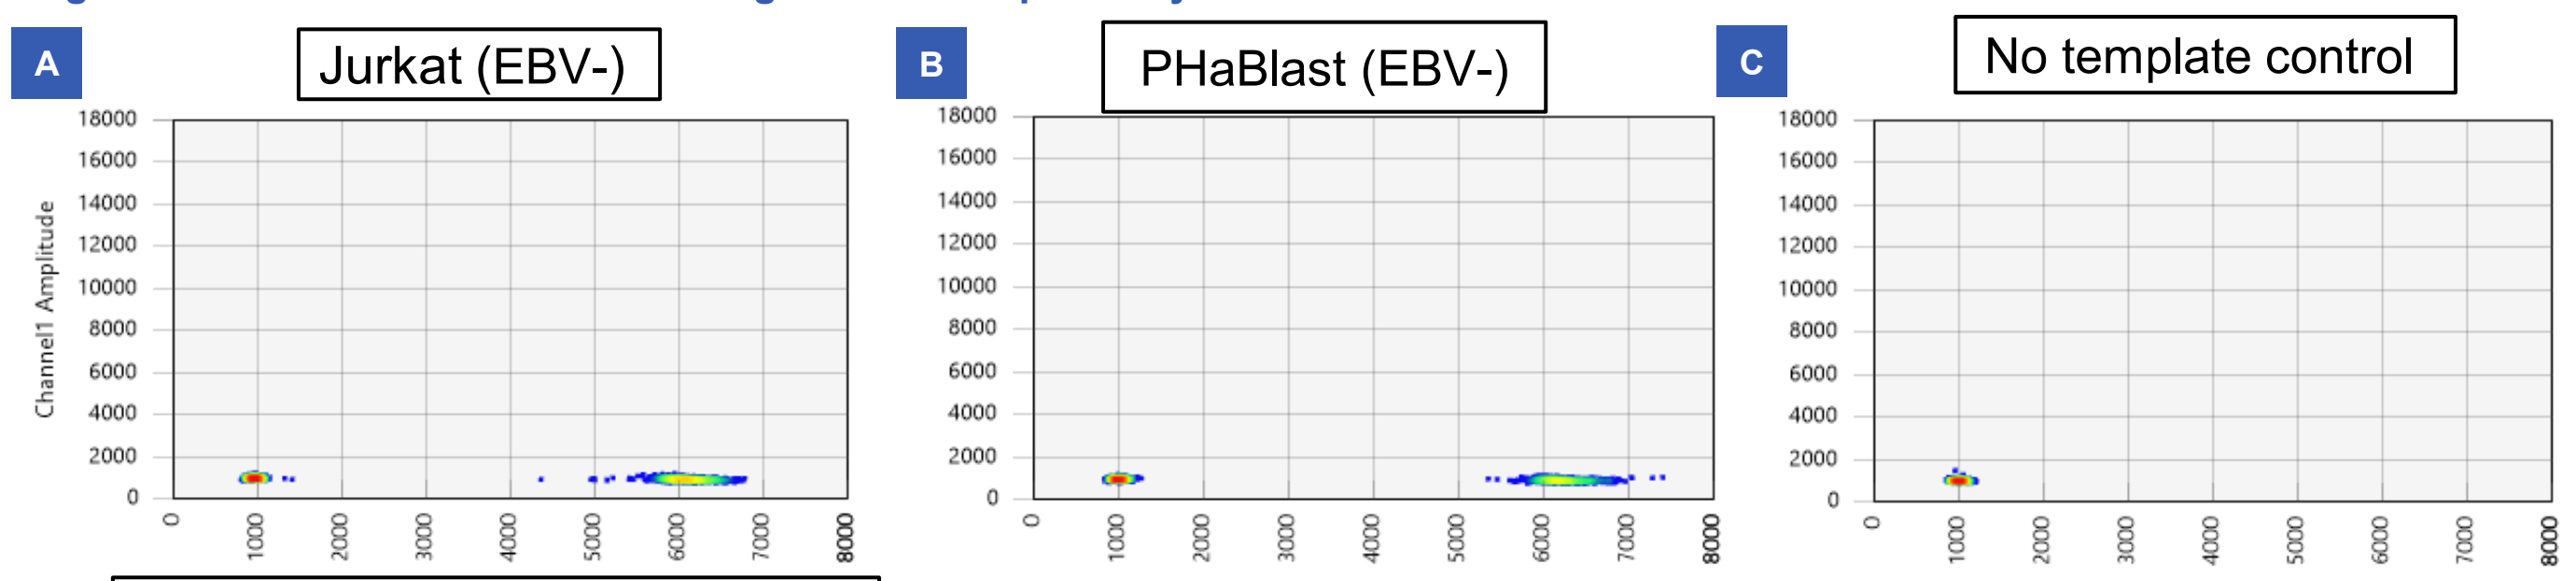
A
Jurkat (EBV-)
B
PHaBlast (EBV-)
C
No template control
Channel1 Amplitude

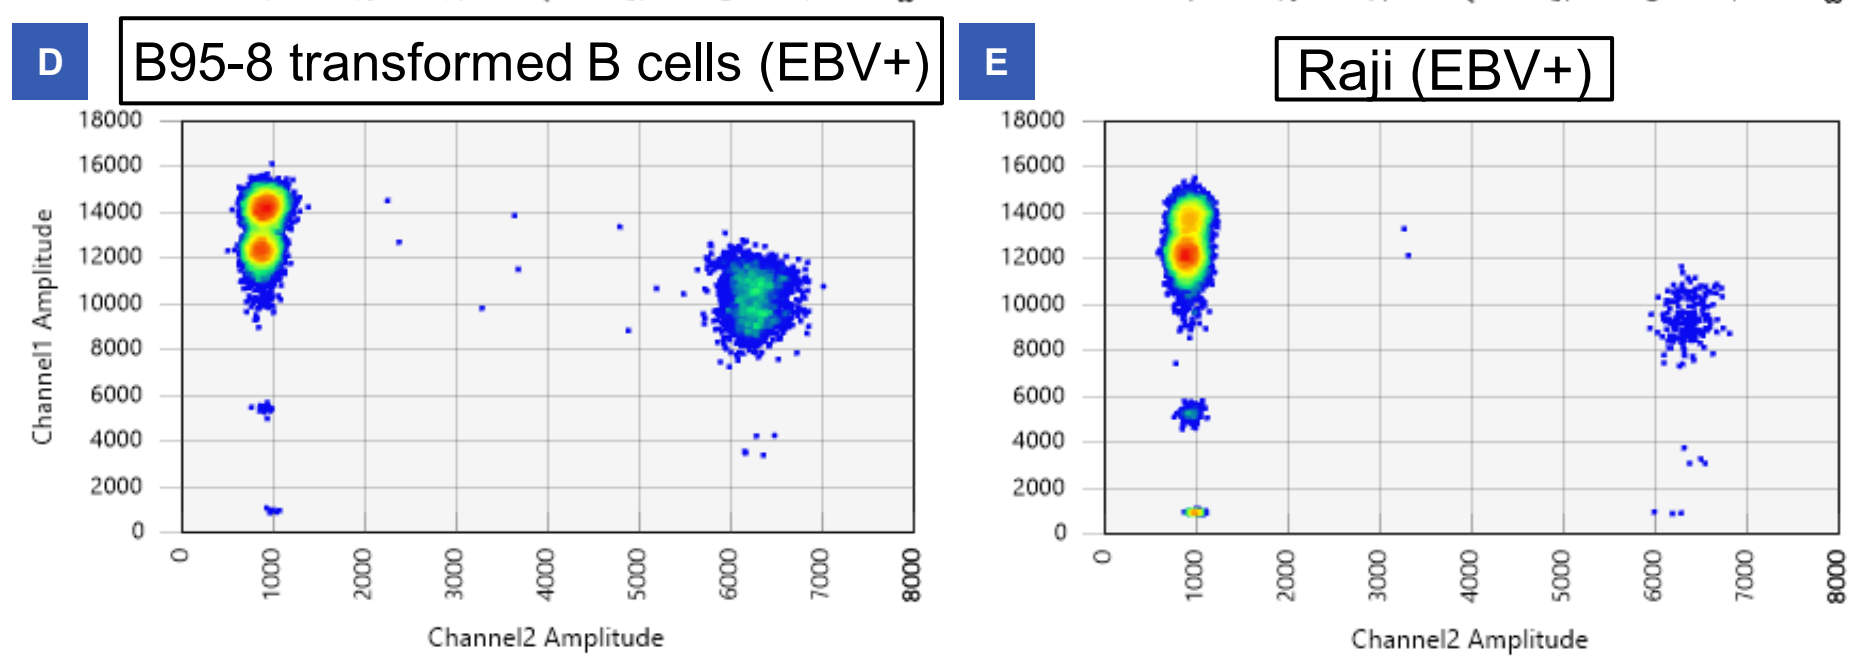
D
B95-8 transformed B cells (EBV+)
E
Raji (EBV+)
Channel1 Amplitude
Channel2 Amplitude

Species	Target	Dye	Amplitude
EBV	BWRF1	FAM	High
EBV	BCLF1	FAM	Low
Human	RPP30	HEX	

A. Jurkat (EBV-) B. PHaBlast (EBV-) C. No template control D. B95-8 transformed B cells (EBV+) E. Raji (EBV+).

Figure 5. Optimization of Analytical Performance

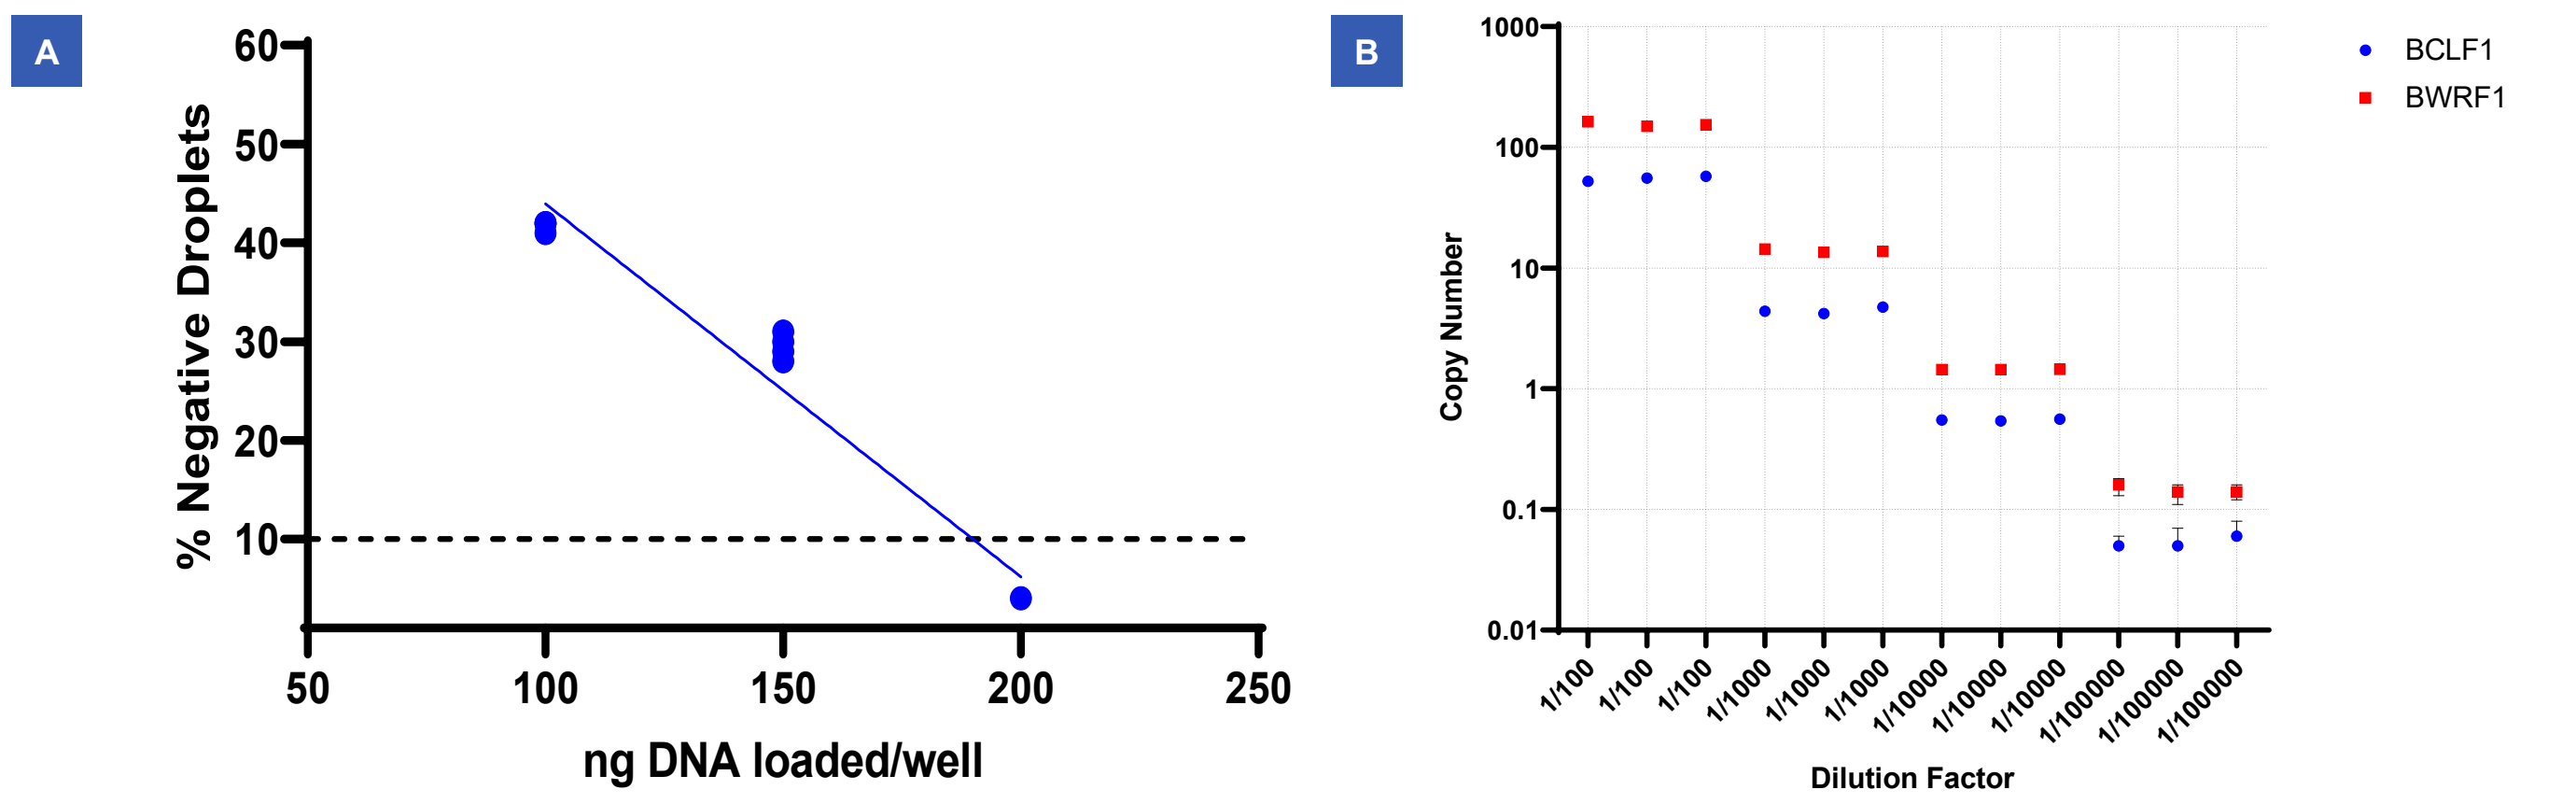
A
% Negative Droplets
ng DNA loaded/well
B
BCLF1
BWRF1
Copy Number
Dilution Factor

A. Defining maximum human genomic DNA load to ensure negative droplet generation for housekeeping gene (HEX) B. 10-fold serial dilution of EBV-positive DNA (RAJI) in EBV-negative DNA (Jurkat).

Figure 6. Developed Assay Allows for Detection Even in Healthy Seropositive Donors

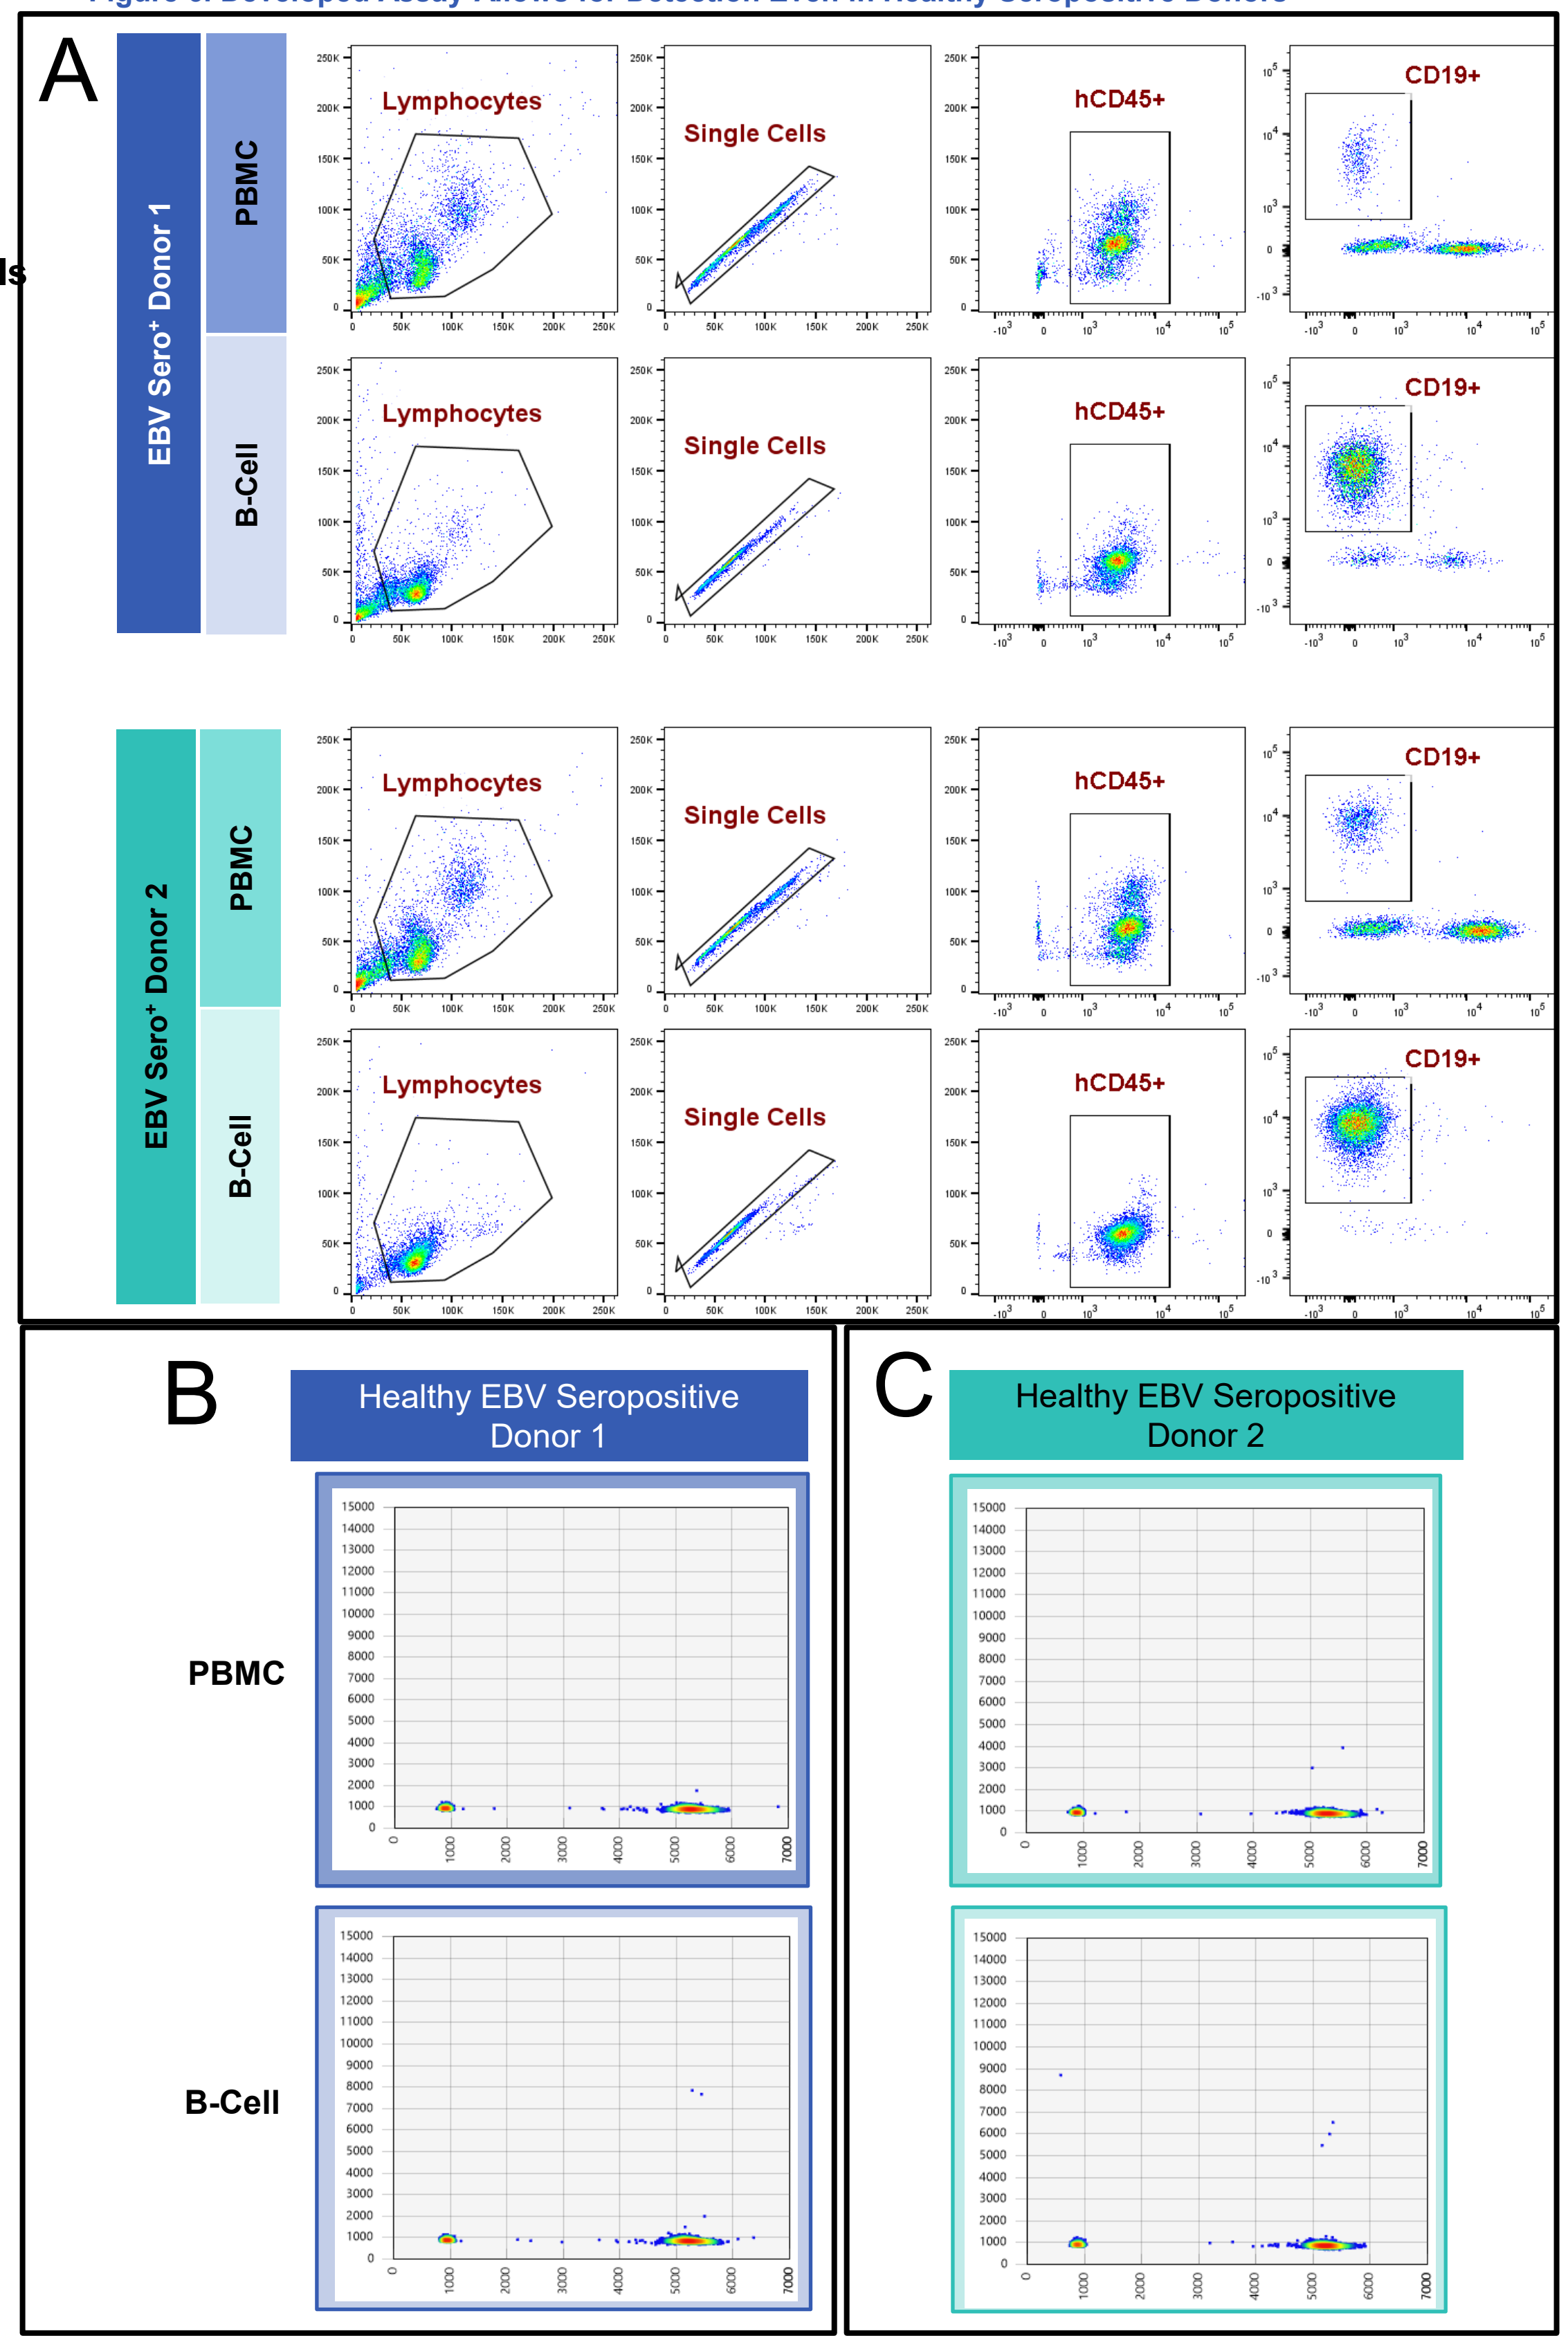
A
EBV Sero+ Donor 1
PBMC
B-Cell
Lymphocytes
Single Cells
hCD45+
CD19+
EBV Sero+ Donor 2
B
Healthy EBV Seropositive Donor 1
C
Healthy EBV Seropositive Donor 2

Figure 6: EBV-DNA can be detected in latently infected healthy EBV seropositive donors. Genomic DNA (gDNA) was isolated from whole PBMCs or from enriched B-cells from two healthy EBV seropositive donors (A). Detection of EBV-DNA is detectable in both whole PBMCs and in enriched B cells isolated from donor 1 (B) and donor 2 (C).

CONCLUSIONS

- **Utilizing a highly sensitive ddPCR approach will allow for absolute quantification of EBV target genes within a sample without use of a standard curve**
- **Upon initial testing, the EBV gene targets are specific to genomic DNA containing EBV DNA**
- **Our optimized ddPCR assay can detect positive droplets containing EBV-positive B-cells of a healthy EBV-seropositive donor**
- **Further validation of this assay will allow its utilization in the EMBOLD study to quantify the presence of EBV-DNA in PBMCs and in the cellular compartment of the CSF from MS patients**
- **Data generated using this technique may be used to support the ATA188 proposed MoA of targeting and eliminating EBV infected cells thought to be involved in the pathogenesis of MS**

REFERENCES

1. Bar-Or A *et al. Trends Mol Med*, 2020; 2. Pender MP *et al. Clin Transl. Immunol*, 2012; 3. Pakpoor J *et al. Mult Scler*, 2012; 4. Dobson R *et al. Neurol Neuroimmunol Neuroinflamm*, 2017; 5. Ruprecht K *et al. ECTRIMS*, 2018; 6. Munger KL *et al. Mult Scler*, 2011; 7. Levin LI *et al. Ann Neurology*, 2010; 8. Ascherio A *et al. Nat Rev Neurol*, 2012; 9. Serafini B *et al. J Exp Med*, 2007; 10. Moreno MA *et al. Neurol Neuroimmunol Neuroinflamm*, 2018; 11. Pender M *et al. Clin & Transl Immunol*, 2017; 12. Pender MP. *Trends Immunol*, 2003; 13. De Leo, Alessandra *et al. Trends in microbiology*, 2020; 14. Kanakry *et al. Blood*, 2016; 15. Rolinski *et al. Blood*, 2014; 16. Kimura *et al. Blood*, 2001; 17. Smatti, Maria K *et al. Frontiers in oncology*, 2018; 18. Tosato, G *et al. The Journal of clinical investigation*, 1984.

Presented at The 29th Annual Meeting of the European Charcot Foundation (ECF 2021), November 14–18, 2021