Phase I open-label extension and imaging data for ATA188, an allogeneic Epstein-Barr virus-targeted multiple sclerosis immunotherapy

Bar-Or A,[1] Pender MP,[2] Hodgkinson SJ,[3,4] Broadley S,[5] Lindsey JW,[6] Ioannides ZA,[2] Bagert B,[7] Gamelin L,[8] Liu E,[8] Ye W,[8] Willmer J,[8] Arnold DL[9,10]

[1]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [2]The University of Queensland, Brisbane, Australia; [3]University of New South Wales, Sydney, Australia; [4]Liverpool Hospital, Liverpool, Australia; [5]Griffith University, Southport, Australia; [6]University of Texas Health Science Center at Houston, Houston, TX, USA; [7]Ochsner Health, New Orleans, LA, USA; [8]Atara Biotherapeutics, South San Francisco, CA, USA; [9]Montreal Neurological Institute, McGill University, Montreal, Canada; [10]NeuroRx Research, Montreal, Canada

BACKGROUND

ATA188 in progressive MS

- Many studies show that EBV infection, particularly in B cells, is strongly involved with the pathogenesis of MS[1–12]
- ATA188 is an investigational, off-the-shelf, allogeneic, T-cell immunotherapy that targets EBV-infected cells
- Sourced and produced from unrelated, EBV-seropositive, immunologically diverse donors, ATA188 is selected for each patient from an existing inventory based on an appropriate HLA restriction and allele profile (**Figure 1**)
- Here, we describe results from Part 1 of a Phase I/II study, which evaluated the safety and potential efficacy of off-the-shelf, allogeneic EBV-targeted T-cell immunotherapy (ATA188) in adults with progressive forms of MS (NCT03283826)
- Efficacy from the 12-month dose-escalation portion of this study was previously reported.[13] In summary, a higher proportion of patients showed SDI with higher doses, which was largely driven by sustained EDSS improvement

Figure 1. ATA188 manufacturing and selection based on HLA restriction

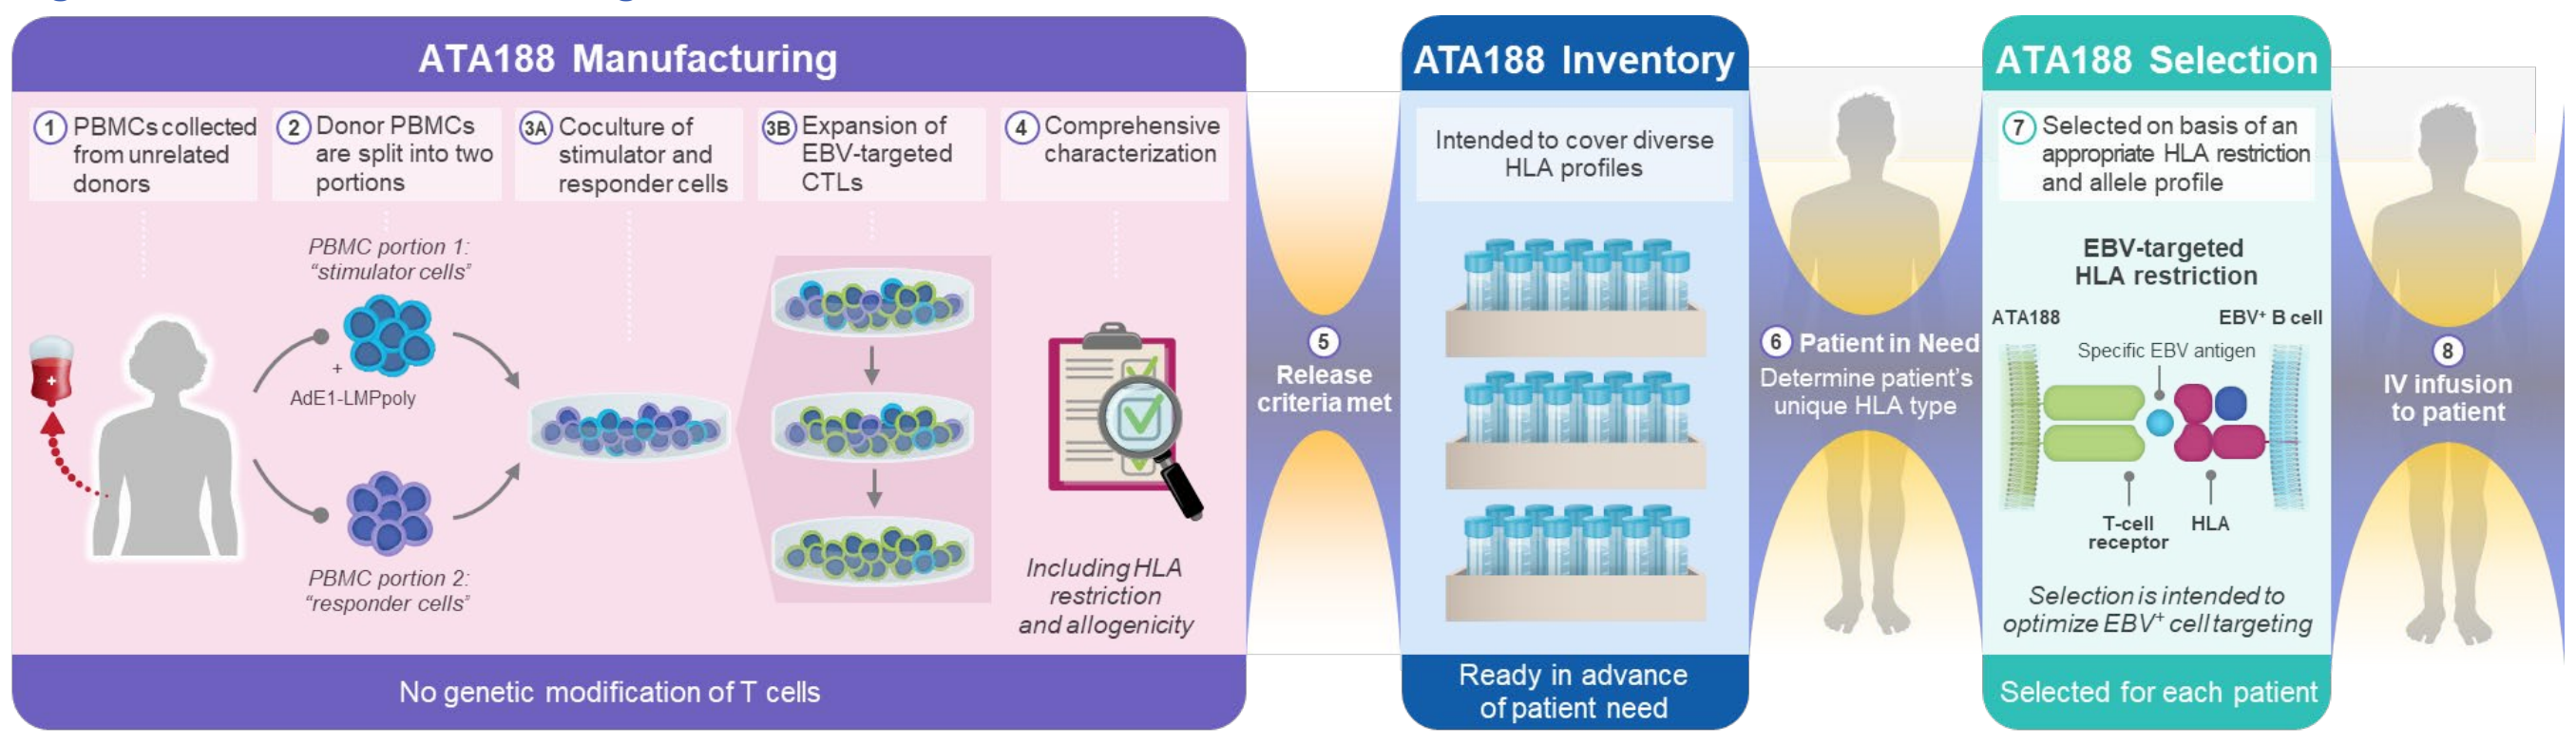
ATA188 Manufacturing
1 PBMCs collected from unrelated donors
2 Donor PBMCs are split into two portions
3A Coculture of stimulator and responder cells
3B Expansion of EBV-targeted CTLs
4 Comprehensive characterization
PBMC portion 1: "stimulator cells"
AdE1-LMPpoly
PBMC portion 2: "responder cells"
Including HLA restriction and allogenicity
No genetic modification of T cells
5 Release criteria met
ATA188 Inventory
Intended to cover diverse HLA profiles
Ready in advance of patient need
6 Patient in Need
Determine patient's unique HLA type
ATA188 Selection
7 Selected on basis of an appropriate HLA restriction and allele profile
EBV-targeted HLA restriction
ATA188
EBV+ B cell
Specific EBV antigen
T-cell receptor
HLA
Selection is intended to optimize EBV+ cell targeting
Selected for each patient
8 IV infusion to patient

METHODS

Study design: Details of Part 1 of this Phase I/II study design were previously reported[14]

- Patients were followed up for 1 year and could participate in a 4-year OLE
- Four cohorts received escalating doses of ATA188 to determine the recommended Part 2 dose

Endpoints: Incidence of AEs and clinically significant changes in laboratory tests, ECGs, and vital signs; identification of the recommended Part 2 dose of ATA188 (primary); and change from baseline in EDSS score

- The following were also assessed: sustained EDSS improvement (**Table 1**), sustained disability improvement (SDI), 25-foot walk time (T25FW), and, as an exploratory endpoint, normalized magnetization transfer ratio (nMTR; assessed by MRI; **Box 1**)
- Percentage of patients with sustained EDSS improvement at 12 months is the primary endpoint of EMBOLD, the Phase II portion of this Phase I/II study

Table 1. Sustained EDSS improvement

Definition	Details
EDSS improvement	Improvement from baseline in EDSS score (minimal clinically significant improvement: –1 for baseline EDSS 3–5; –0.5 for baseline EDSS 5.5–7.0)
Sustained EDSS improvement at 6 months; 12 months	EDSS improvement at 3 months and confirmed at 6 months; EDSS improvement at 6 months and confirmed at 12 months
Sustained EDSS improvement in the OLE	EDSS improvement at any two consecutive visits (eg at 12 months and confirmed at 15 months)

As previously described,[13,14] SDI is defined by sustained improvements in EDSS score (as shown above) and/or T25FW (minimal clinically significant improvement –20%)

Box 1. Normalized magnetization transfer ratio

Changes in nMTR are a marker of changes in myelin density

- An increase in nMTR can reflect remyelination, and a decrease in nMTR can reflect demyelination
- There is an association between nMTR signal and disability change as measured by EDSS;[15] as such, **nMTR may be a radiologic biomarker of EDSS improvement**

In this study, nMTR was measured in two compartments:

- **Unenhancing T2 lesions** – these are mainly chronic but could include some subacute regions
- **Normal-appearing brain tissue** – this includes all tissue (white and grey matter) that is not lesion

To minimize potential site-to-site variability related to scanner differences, a previously published normalization method developed for this specific application was used[16]

RESULTS – PATIENT DISPOSITION

Figure 2. Summary of patients evaluated in the Part 1 dose-escalation portion and OLE

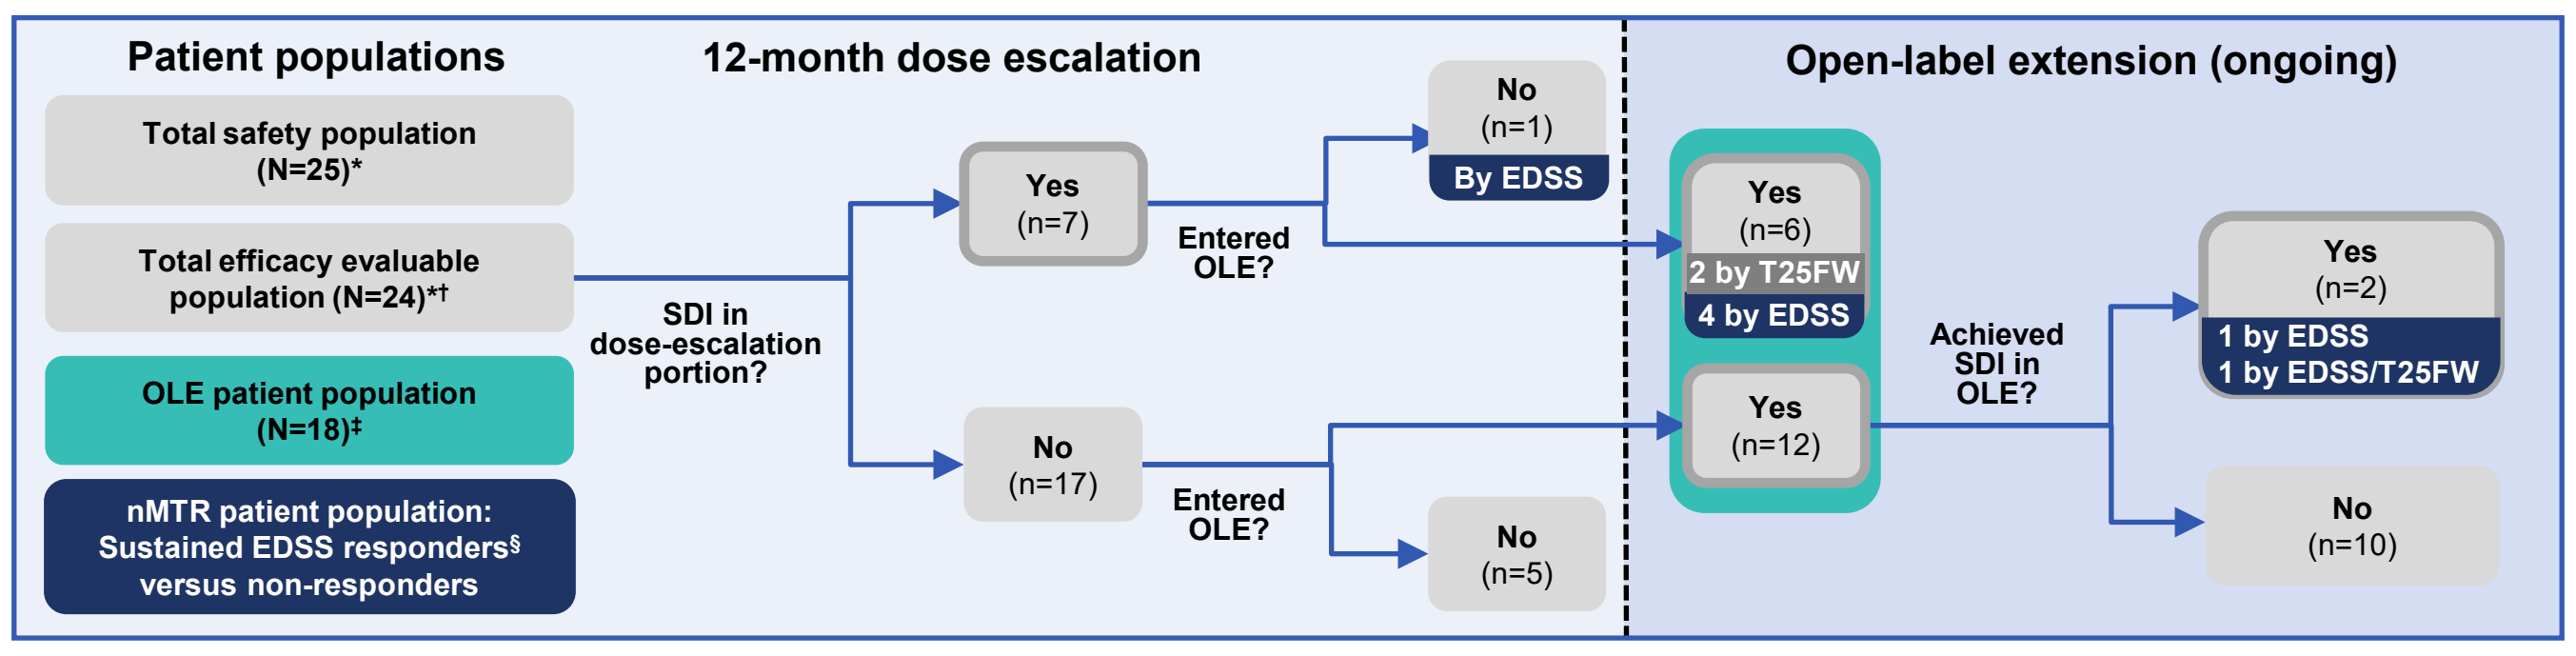
Patient populations
Total safety population (N=25)*
Total efficacy evaluable population (N=24)*†
OLE patient population (N=18)‡
nMTR patient population: Sustained EDSS responders§ versus non-responders
12-month dose escalation
SDI in dose-escalation portion?
Yes (n=7)
No (n=17)
Entered OLE?
No (n=1) By EDSS
No (n=5)
Open-label extension (ongoing)
Yes (n=6) 2 by T25FW 4 by EDSS
Yes (n=12)
Achieved SDI in OLE?
Yes (n=2) 1 by EDSS 1 by EDSS/T25FW
No (n=10)

*In patients receiving ≥1 dose of ATA188; one patient who had treatment-related MS relapse 7 days after dosing in the setting of ongoing URTI symptoms and possible dental infection discontinued the study and was not evaluated for efficacy (only safety); this patient was replaced with a new patient who was evaluated for both safety and efficacy; †24 patients were evaluated for efficacy at 6 months and 23 patients were evaluated at 12 months (one patient in Cohort 3 was withdrawn, moved out of the country, and lost to 12-month follow-up); ‡In patients receiving all six doses in the initial 12-month dose-escalation portion of the study and followed up for up to 39 months as of the August 2021 data cut-off; §Patients who achieved sustained EDSS improvement either during the initial 12-month portion or in the OLE were considered sustained EDSS responders for the nMTR analysis; nMTR was assessed based on MRI readings taken at 6 and 12 months of the dose-escalation portion of the study

EFFICACY – OPEN-LABEL EXTENSION

As of August 2021, OLE data were available for 18 patients followed for up to 39 months (Tables 2 and 3)

- 9 patients achieved SDI either during the initial 12 months or the OLE, 7 of whom did so via sustained EDSS improvement.
 - 7 patients achieved SDI during the initial 12 months, 6 of whom continued into the OLE (**Figure 2** and **Table 2**)
 - The patient with SDI in the first 12 months who did not enroll in the OLE is included in **Table 2** for completeness
 - An additional 2 patients who did not meet SDI criteria during the initial 12 months met them during the OLE
- Of the 8 patients enrolled in the OLE who achieved SDI at any point in the study (**Figure 2**), 7 maintained SDI at all subsequent time points evaluated and 1 did not (**Table 2**). The median time over which SDI was sustained in these 8 patients was 18 months (range 0.03–27.0 months).
- Patients who did not meet SDI criteria but who entered the OLE (n=10) are shown in **Table 3**

Table 2. EDSS and T25FW results among patients in Cohorts 1–4 who met SDI criteria within the first 12 months and/or during the OLE*

Cohort	Patient	SDI First Achieved	SDI based on EDSS and/or T25FW	Scale	Baseline	3 months	6 months	12 months	15 months	18 months	21 months	24 months	27 months	30 months	33 months
1 (5 x 10⁶ cells)	A (101-003)	6 months	EDSS	EDSS score	4.5	3.0	3.0	3.0	Patient A did not enroll in OLE						
				ΔT25FW	–	–3.0%	+15.2%	–3.0%							
	H (103-001)†	24 months	EDSS, T25FW	EDSS score	5.5	5.5	5.5	5.5	–	–	5.0	5.0	3.5	3.0	3.5
				ΔT25FW	–	–11.0%	–21.5%	–19.2%	–	–	–30.8%	–41.3%	–38.4%	–39.0%	–27.3%
2 (1 x 10⁷ cells)	B (103-010)	6 months	T25FW	EDSS score	6.0	6.0	6.0	6.0	6.0	6.0	6.0	6.0	6.0	–	6.0
				ΔT25FW	–	–21.1%	–37.2%	–37.8%	–31.7%	–30.0%	–29.4%	–30.0%	–37.8%	–	–36.1%
3 (2 x 10⁷ cells)	C (101-004)	12 months	EDSS	EDSS score	6.0	6.0	5.0	5.0	5.0	5.0	4.5	4.5	4.5	–	–
				ΔT25FW	–	–8.2%	–10.2%	–	–17.7%	–7.5%	–8.2%	–16.3%	–20.4%	–	–
	D (103-007)	6 months	T25FW	EDSS score	6.0	6.0	6.0	6.0	6.0	6.0	6.0	6.0	–	–	–
				ΔT25FW	–	–34.8%	–40.9%	–58.2%	–49.0%	–58.4%	–46.0%	–44.0%	–	–	–
	E (103-008)	6 months	EDSS	EDSS score	5.5	3.5	3.5	3.5	3.0	4.0	3.0	4.5	–	3.0	–
				ΔT25FW	–	–10.8%	–13.3%	–0.8%	–19.2%	–3.3%	–5.8%	–6.7%	–	–5.8%	–
4 (4 x 10⁷ cells)	F (210-001)	6 months	EDSS	EDSS score	6.5	6.0	6.0	6.0	6.0	6.0	6.0	6.0	–	–	–
				ΔT25FW	–	–0.9%	–11.1%	–3.5%	+52.5%	+2.8%	+19.3%	+80.1%	–	–	–
	G (210-003)	6 months	EDSS	EDSS score	6.0	5.5	5.0	4.5	5.0	4.5	4.5	4.5	–	–	–
				ΔT25FW	–	+14.5%	–8.1%	–16.1%	–8.1%	–9.7%	–5.6%	–17.7%	–	–	–
	K (210-006)	15 months	EDSS	EDSS score	5.5	5.5	5.5	4.5	4.5	5.5	Patient K had a relapse at 18 months and decided to discontinue the study to try an alternative therapy				
				ΔT25FW	–	+15.2%	–12.9%	+17.4%	+9.1%	+8.3%					

Table 3. EDSS and T25FW results among patients in Cohorts 1–4 with OLE data – patients without SDI

Cohort	Patient	Scale	Baseline	3 months	6 months	12 months	15 months	18 months	21 months	24 months	27 months	30 months	33 months	36 months	39 months
1 (5 x 10⁶ cells)	P (101-005)†	EDSS score	6.0	6.0	6.0	6.0	–	–	–	–	6.0	6.0	6.0	6.0	–
		ΔT25FW	–	+16.7%	+16.7%	+45.8%	–	–	–	–	+30.0%	+21.7%	+41.7%	+21.7%	+57.5%
	Q (101-006)†	EDSS score	6.0	6.0	6.0	6.0	–	–	–	–	–	6.0	6.0	6.0	6.0
		ΔT25FW	–	+28.3%	+12.4%	+53.1%	–	–	–	–	–	+123.4%	+234.5%	+200.7%	+185.5%
2 (1 x 10⁷ cells)	L (201-003)†	EDSS score	4.0	4.0	4.0	4.0	–	4.0	4.0	4.0	3.5	3.5	–	–	–
		ΔT25FW	–	–20.5%	–19.9%	–17.2%	–	–12.6%	–14.6%	–21.2%	–17.9%	–12.6%	–	–	–
	S (101-008)†	EDSS score	6.5	6.5	6.5	6.5	–	–	–	–	7.5	7.5	7.5	–	–
		ΔT25FW	–	–20.1%	+3.6%	+94.4%	–	–	–	–	–‡	–‡	–‡	–	–
	R (102-002)†	EDSS score	6.5	6.5	6.5	6.5	–	–	6.5	6.5	6.5	–	–	–	–
		ΔT25FW	–	+30.7%	+59.0%	+65.1%	–	–	+177.7%	+413.9%	+379.5%	–	–	–	–
3 (2 x 10⁷ cells)	I (101-002)	EDSS score	6.5	6.5	6.5	6.5	6.0	6.5	6.5	6.5	–	–	–	–	–
		ΔT25FW	–	+19.3%	+43.9%	+24.7%	+47.5%	+48.0%	+13.5%	+63.7%	–	–	–	–	–
	J (103-006)	EDSS score	4.5	4.5	6.0	6.0	6.0	6.0	6.0	6.0	–	–	–	–	–
		ΔT25FW	–	–5.3%	–12.8%	+22.6%	+9.0%	–15.0%	–15.0%	–11.3%	–	–	–	–	–
4 (4 x 10⁷ cells)	M (101-011)	EDSS score	6.0	6.0	6.0	6.0	6.0	6.0	6.5	6.0	–	–	–	–	–
		ΔT25FW	–	+24.4%	+10.7%	–7.6%	+9.6%	+16.8%	+24.9%	+18.8%	–	–	–	–	–
	N (102-004)	EDSS score	6.0	6.0	6.0	6.5	7.0	7.0	6.0	7.5	–	–	–	–	–
		ΔT25FW	–	+3.4%	+31.2%	+65.4%	–‡	–‡	–‡	–‡	–	–	–	–	–
	O (210-002)	EDSS score	6.0	5.5	6.0	6.0	6.0	6.0	6.0	6.0	–	–	–	–	–
		ΔT25FW	–	+2.9%	–3.3%	–21.9%	+31.4%	+0.8%	0.0%	+21.1%	–	–	–	–	–

Clinically significant improvement; Trend for improvement/stable; Clinically significant decline; Trend for decline; Redosed for OLE Year 2 – Cohort 3 dose; Redosed for OLE Year 3 – Cohort 4 dose‡

Minimal clinically significant improvement: EDSS score –1 for baseline EDSS 3–5, –0.5 for baseline EDSS 5.5–7.0; T25FWT –20%. Clinically significant decline is defined as the same magnitude as improvement but in the opposite direction. *Columns for 36 months and 39 months are not shown as there are no data available for the patients who achieved SDI as of August 2021 at these timepoints; †Following the 12-month assessment, the patient had a treatment gap before redosing for the OLE and did not undergo any scheduled assessments during the interim period; ‡Patient was unable to complete the test at this time point because of physical limitations; therefore, the result was recorded as 'decline' with no associated numerical value. ‡Subjects 101-002,103-001,103-006,103-007,103-008,103-010 received cohort 3 dose for their year 3 redosing.

EFFICACY – NORMALIZED MAGNETIZATION TRANSFER RATIO

Changes in nMTR from baseline for unenhancing T2 lesions and normal-appearing brain tissue were assessed at 6 and 12 months in patients who achieved sustained EDSS improvement at any point in the study versus those who did not (Figure 3)

- Patients achieving sustained EDSS improvement at any time (versus those who did not):
 - Showed a significant increase from baseline in nMTR for unenhancing T2 lesions at 12-months (**Figure 3B**)
 - Showed a greater increase from baseline in nMTR for unenhancing T2 lesions at 6-months (**Figure 3A**)
 - Showed a greater increase from baseline in nMTR for normal-appearing brain tissue at 12-months (**Figures 3D**)
 - This trend appeared to occur in patients with either PPMS or SPMS (**Figures 3A, 3B and 3D**)
- Compared to baseline, nMTR at 12 months for unenhanced T2 lesions and normal-appearing brain tissue in patients with sustained EDSS improvement increased (median change of 0.134 and 0.082, respectively), whereas nMTR in those patients without sustained EDSS improvement remained unchanged (median change of –0.030 and 0.005, respectively).
- In general, a trend supporting a relationship between improvement in nMTR signal and decrease in EDSS score (improvement in disability) was observed (**Figure 4**)

Figure 3. nMTR at 6 and 12 months in patients achieving and not achieving sustained EDSS improvement at any time (including OLE)*

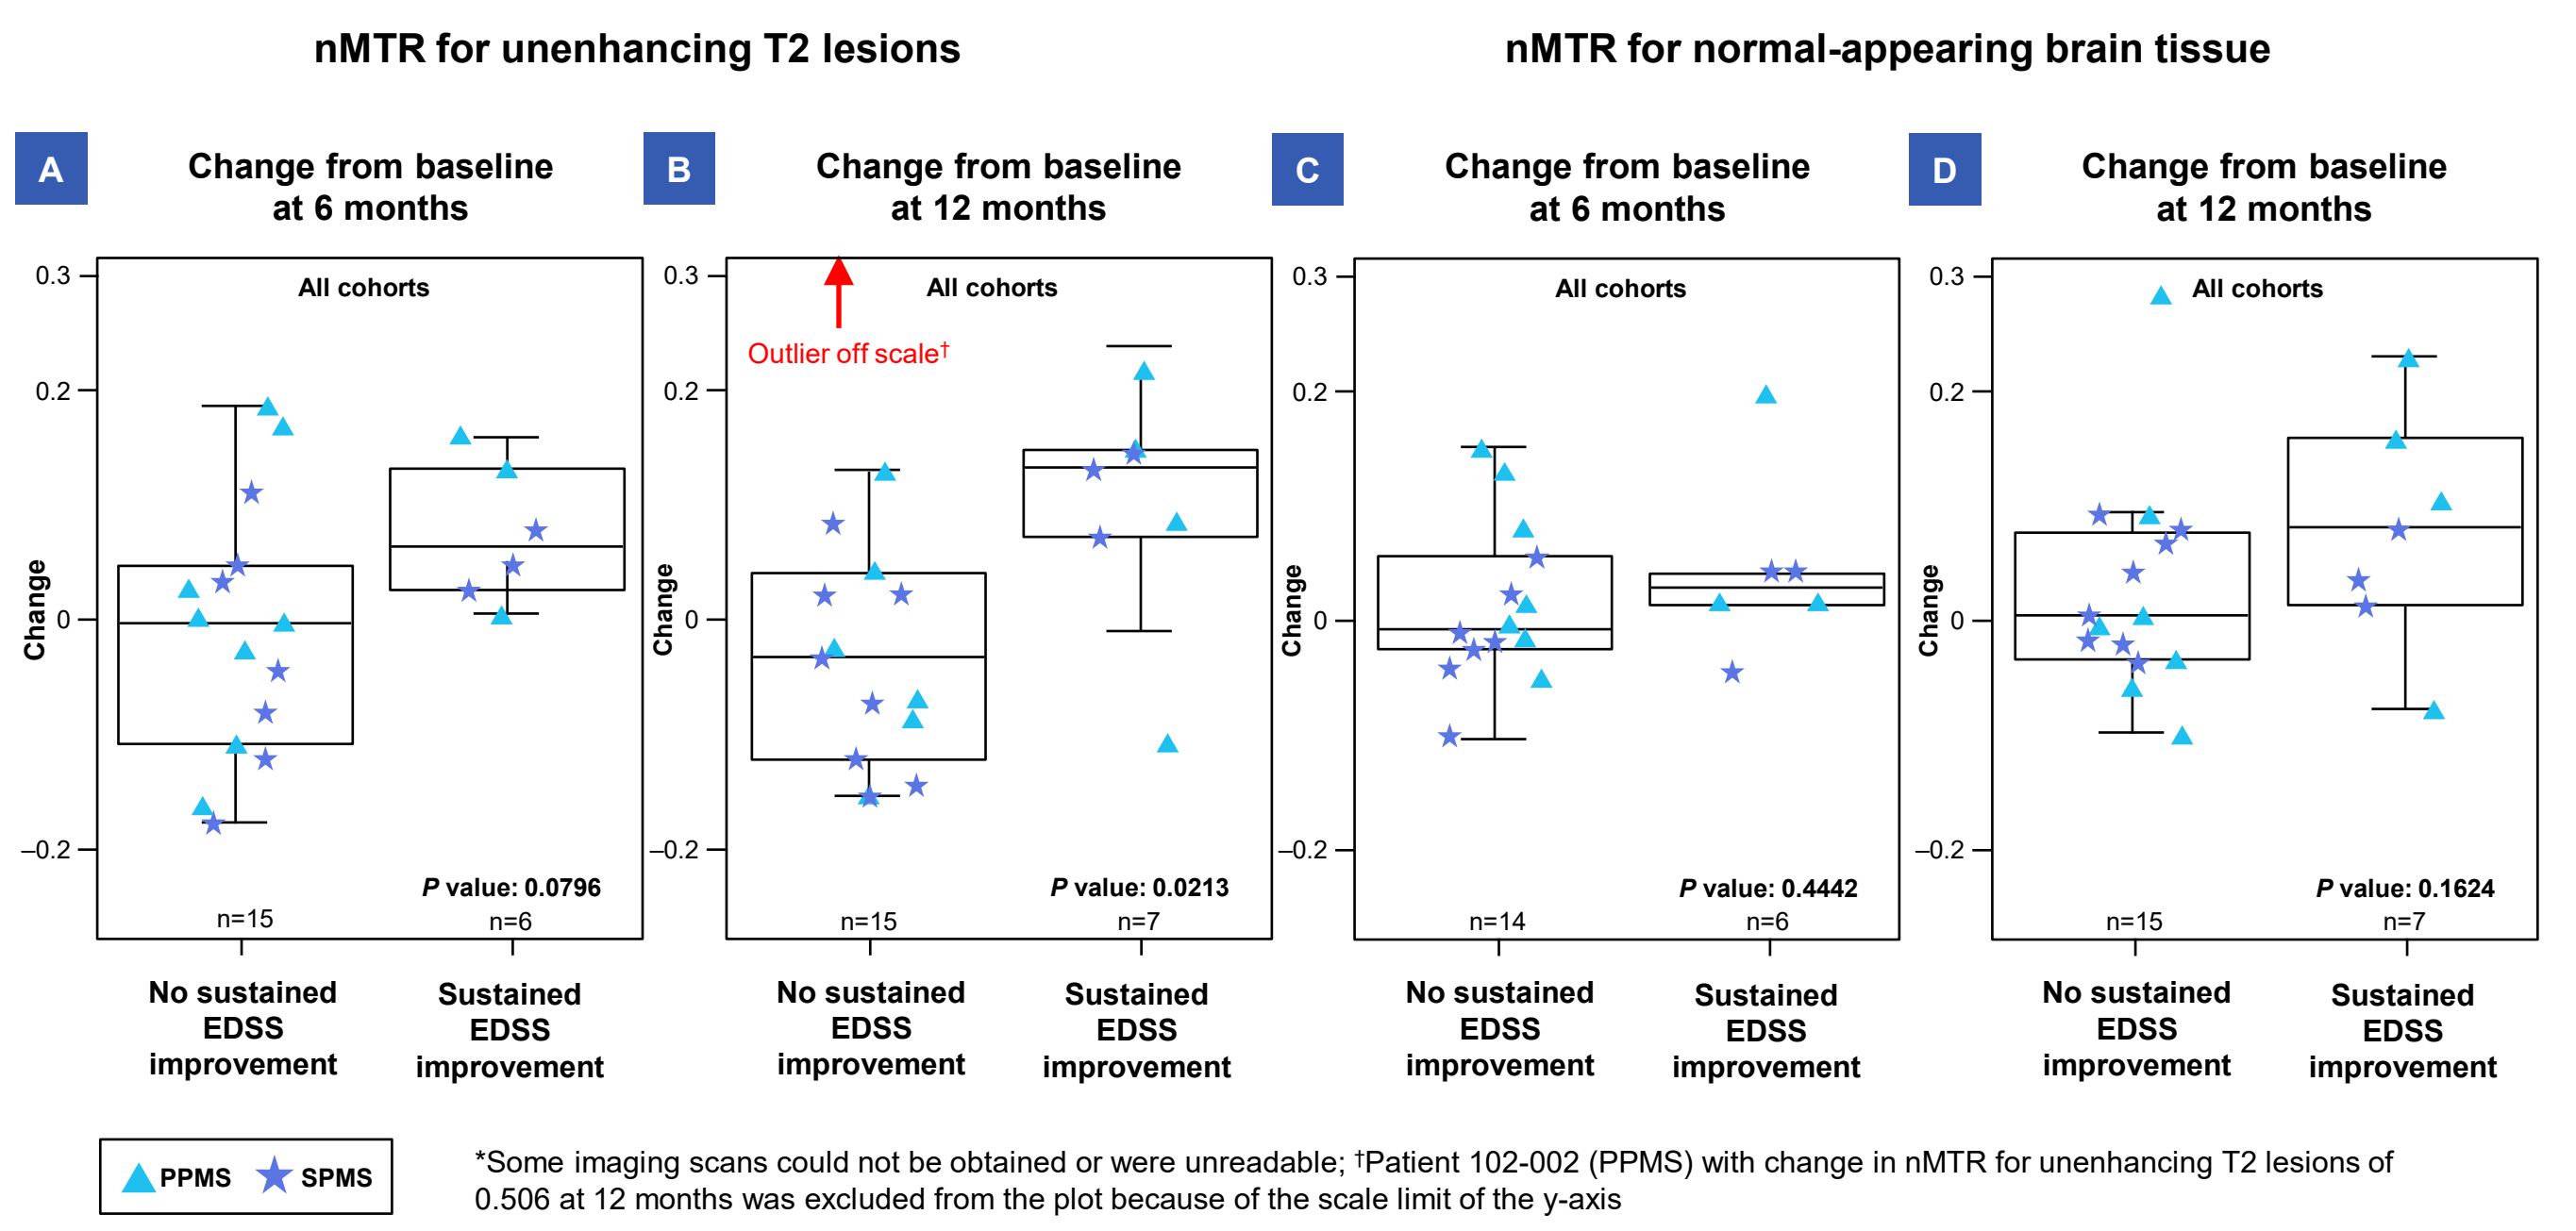
nMTR for unenhancing T2 lesions
nMTR for normal-appearing brain tissue
A Change from baseline at 6 months
B Change from baseline at 12 months
C Change from baseline at 6 months
D Change from baseline at 12 months
All cohorts
Outlier off scale†
Change
P value: 0.0796
P value: 0.0213
P value: 0.4442
P value: 0.1624
n=15 n=6
n=15 n=7
n=14 n=6
n=15 n=7
No sustained EDSS improvement
Sustained EDSS improvement
PPMS SPMS

*Some imaging scans could not be obtained or were unreadable; †Patient 102-002 (PPMS) with change in nMTR for unenhancing T2 lesions of 0.506 at 12 months was excluded from the plot because of the scale limit of the y-axis

Figure 4. nMTR versus EDSS score at 6 and 12 months*

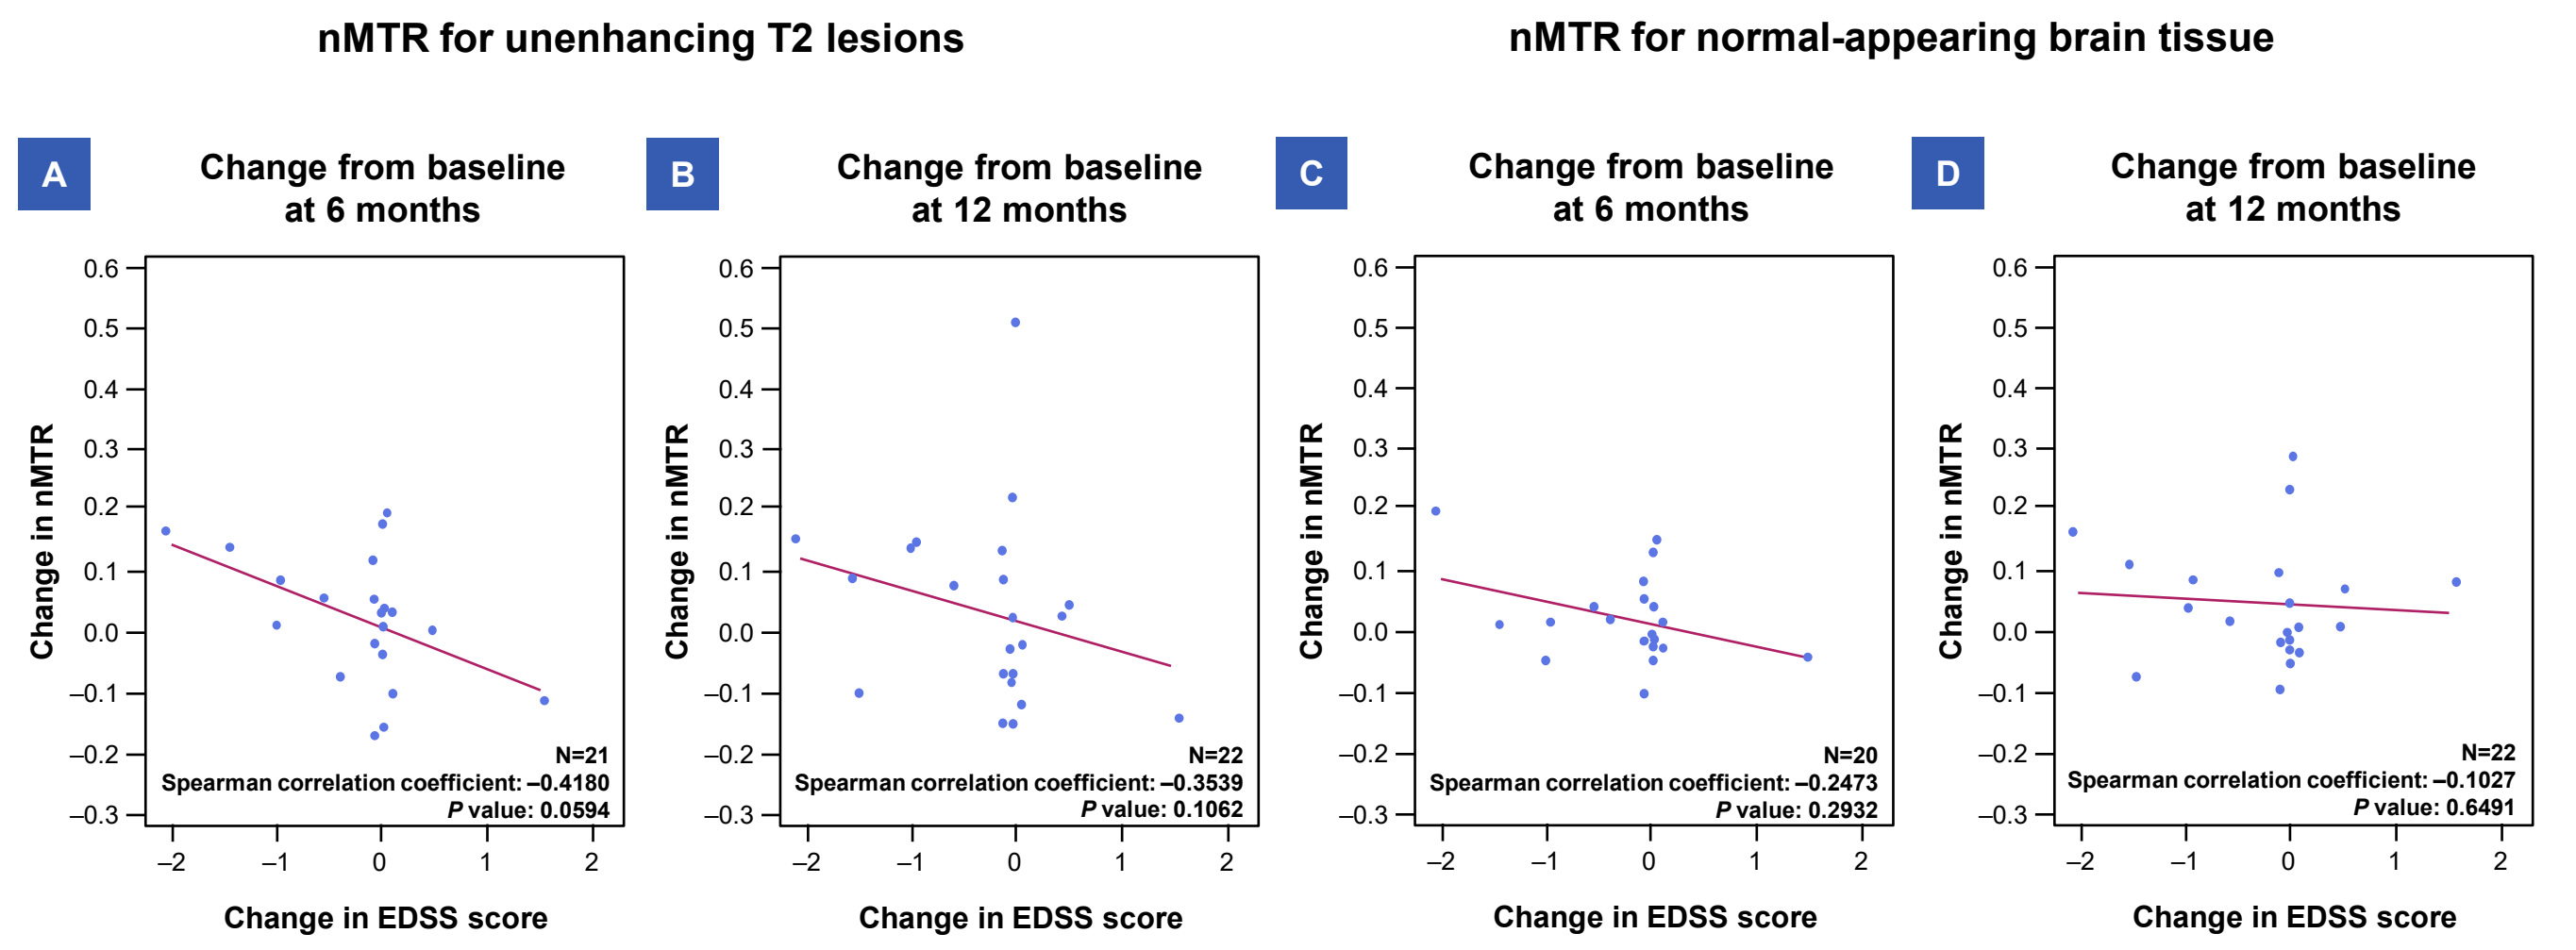
nMTR for unenhancing T2 lesions
nMTR for normal-appearing brain tissue
A Change from baseline at 6 months
B Change from baseline at 12 months
C Change from baseline at 6 months
D Change from baseline at 12 months
Change in nMTR
Change in EDSS score
N=21 Spearman correlation coefficient: –0.4180 P value: 0.0594
N=22 Spearman correlation coefficient: –0.3539 P value: 0.1062
N=20 Spearman correlation coefficient: –0.2473 P value: 0.2932
N=22 Spearman correlation coefficient: –0.1027 P value: 0.6491

*Some imaging scans could not be obtained or were unreadable

SAFETY

25 patients received ≥1 dose of ATA188 and were evaluated for safety

As of August 2021, inclusive of the OLE, in which patients were followed up for up to 39 months:

- No Grade >3 events, dose-limiting toxicities, cytokine-release syndrome, or graft-versus-host disease were observed
- Three treatment-emergent SAEs were reported, as follows:
 - One patient in Cohort 4 had a Grade 3 SAE of MS relapse, reported as possibly related to treatment, 7 days after dosing in the setting of ongoing URTI symptoms and possible dental infection
 - One patient in Cohort 3 had a Grade 2 SAE of muscle spasticity, reported as unrelated to treatment
 - One patient in Cohort 4 had a Grade 2 SAE of fall, reported as unrelated to treatment [occurred during OLE]

CONCLUSIONS

- **Updated OLE results in patients with progressive forms of MS treated with ATA188 followed for up to 39 months, as well as new normalized magnetization transfer ratio data showed the following:**
 - **Sustained clinical benefit:** 7 of the 8 patients who enrolled in the OLE and achieved SDI at any time point maintained SDI at all future time points. In the majority, SDI was driven by sustained EDSS improvement.
 - **Evidence of Possible Remyelination:** Patients who achieved sustained EDSS improvement at any time in the study (versus those who did not) showed greater increases in nMTR from baseline at 12 months, which may be suggestive of remyelination. In general, an increase in nMTR was associated with improvement in EDSS scores.
 - **Safety and tolerability:** Preliminary data suggest that ATA188 is safe and well tolerated.
- **These data suggest that following ATA188 treatment, patients may achieve SDI, and specifically sustained EDSS improvement, at a higher rate and longer duration than would be expected based on the natural history of progressive MS. The nMTR data provide evidence that structural changes suggestive of remyelination may be driving such prolonged sustained improvement.**
- **Although encouraging, these results need to be confirmed within a randomized, placebo-controlled study.**

The Phase II RCT portion of this study, EMBOLD (NCT03283826), is ongoing and currently enrolling

ABBREVIATIONS

ΔT25FW, change in T25FW from baseline; AdE1-LMPpoly, recombinant adenoviral vector encoding EBNA1, LMP1, and LMP2A; AE, adverse event; CTL, cytotoxic T lymphocyte; EBNA1, EBV nuclear antigen 1; EBV, Epstein–Barr virus; ECG, electrocardiogram; EDSS, Expanded Disability Status Scale; HLA, human leukocyte antigen; LMP1, latent membrane protein 1; LMP2A, latent membrane protein 2A; MRI, magnetic resonance imaging; MS, multiple sclerosis; nMTR, normalized magnetization transfer ratio; OLE, open-label extension; PBMC, peripheral blood mononuclear cell; PPMS, primary progressive MS; RCT, randomized controlled trial; SAE, serious adverse event; SDI, sustained disability improvement; SPMS, secondary progressive MS; T25FW, 25-foot walk time; URTI, upper respiratory tract infection

REFERENCES

1. Bar-Or A *et al. Trends Mol Med* 2020;26:296–310; 2. Pender MP *et al. Clin Transl Immunol* 2014;3:e27; 3. Pakpoor J *et al. Mult Scler* 2013;19:162–6; 4. Dobson R *et al. Neurol Neuroimmunol Neuroinflamm* 2017;4:e318; 5. Ruprecht K *et al.* ECTRIMS 2018; 6. Munger KL *et al. Mult Scler* 2011;17:1185–93; 7. Levin LI *et al. Ann Neurol* 2010;67:824–30; 8. Ascherio A *et al. Nat Rev Neurol* 2012;8:602–12; 9. Serafini B *et al. J Exp Med* 2007;204:2899–912; 10. Moreno MA *et al. Neurol Neuroimmunol Neuroinflamm* 2018;5:e466; 11. Pender MP *et al. Clin Transl Immunol* 2017;6:e126; 12. Pender MP. *Trends Immunol* 2003;24:584–8; 13. Pender MP *et al.* ECF 2020; 14. Pender MP *et al. Eur J Neurol* 2020;27(Suppl 1):1268–307 (abst LB130); 15. Fox RJ *et al. Neurol Clin* 2011;29:357–80; 16. 4. Brown RA *et al. Proc Intl Soc Mag Reson Med* 2011;19

DISCLOSURES

- This study is sponsored and funded by Atara Biotherapeutics (NCT03283826).
- Medical writing assistance was provided by Cindy Chiu, PhD from AMICULUM Ltd, funded by Atara Biotherapeutics
- **Amit Bar-Or** has received consulting fees and is a speaker in meetings sponsored by Janssen/Actelion, Atara Biotherapeutics, Biogen Idec, Celgene/Receptos, Roche/Genentech, MAPI, Medimmune, Merck/EMD Serono, Novartis, and Sanofi-Genzyme. He has received grant support from Janssen/Actelion, Atara Biotherapeutics, Biogen Idec, Celgene/Receptos, Roche/Genentech, MAPI, Medimmune, Merck/EMD Serono, Novartis, and Sanofi-Genzyme. **Michael P Pender** reports grants from MS Queensland, a research grant from Atara Biotherapeutics, clinical trial support from Atara Biotherapeutics, personal consulting fees from Atara Biotherapeutics, travel support from Atara Biotherapeutics, and membership of the Scientific Advisory Board of Atara Biotherapeutics. **Suzanne Hodgkinson** reports clinical trial support from Atara Biotherapeutics and honoraria for advisory boards from Biogen, Novartis, Merck, Roche, and Sanofi. **Simon Broadley** reports clinical trial support through Griffith University from Atara Biotherapeutics and has received travel sponsorship to attend international scientific meetings, speaker's honoraria, and fees for advisory board attendance from Bayer-Schering, Biogen Idec, Merck, Novartis, Sanofi-Genzyme, and TEVA. **J William Lindsey** has received personal compensation for speaking or consulting for EMD Serono, Celgene, Mapi Pharmaceuticals, Banner Life Sciences, TG Therapeutics, Genentech, and Genzyme, is participating in clinical trials funded by Genentech, Biogen, Atara Biotherapeutics, EMD Serono, and AbbVie, and has received research funding from the National Multiple Sclerosis Society and Genentech. **Zara A Ioannides** reports grants from MS Queensland and clinical trial support from Atara Biotherapeutics. **Bridget Bagert** has received grant support from Ochsner Health, the National Multiple Sclerosis Society, and the Multiple Sclerosis Foundation. She participates in clinical trials supported by Atara Biotherapeutics, Genentech, Abbvie, Biogen, Sanofi-Genzyme, and GW Pharmaceuticals. **Laurence Gamelin, Emily Liu, Wei Ye,** and **Jonathan Willmer** are employees and stockholders of Atara. **Douglas Arnold** reports consulting fees from Albert Charitable Trust, Alexion Pharma, Biogen, Celgene, Frequency Therapeutics, Genentech, Med-Ex Learning, Merck, Novartis, Population Council, Receptos, Roche, and Sanofi-Aventis, grants from Biogen, Immunotec, and Novartis, and an equity interest in NeuroRx

Presented at The 29th Annual Meeting of the European Charcot Foundation (ECF 2021), November 14–18, 2021